Changes in Plasma and Urinary Metabolites After Elamipretide in Barth Syndrome Patients: Analyses from the TAZPOWER Study

David A. Brown[1], Hilary J. Vernon[2]

[1]Stealth BioTherapeutics, Newton, MA; [2]Johns Hopkins University, Baltimore, MD

INTRODUCTION

- Elamipretide localizes to the inner mitochondrial membrane and associates with cardiolipin to improve mitochondrial membrane stability and ATP production
- Barth syndrome (BTHS) is an inherited disorder that results from pathogenic mutations in *TAZ*, the gene responsible for encoding taffazin, an enzyme that remodels the mitochondrial phospholipid cardiolipin
- BTHS is characterized by heart and skeletal muscle myopathy, growth delay, and neutropenia
- There are no approved therapies for BTHS and a paucity of reliable biomarkers with which to understand disease progression and appropriate treatment in this patient population
- Increased plasma levels of short- and medium-chain acylcarnitines (C2 to C16) are frequently observed biomarkers of mitochondrial dysfunction, and are commonly seen across myopathic diseases and muscle wasting
- TAZPOWER clinical trial evaluated the effects of elamipretide on functional outcomes for patients with BTHS[1]

OBJECTIVE

- Exploratory targeted metabolomic analyses were performed to assess changes in plasma and urinary metabolites for patients with BTHS receiving elamipretide and placebo in TAZPOWER

METHODS

TAZPOWER Study Design

- TAZPOWER (NCT03098797) was a 28-week, randomized, double-blind, placebo-controlled crossover assessment of the safety and efficacy of elamipretide in patients with BTHS (**Figure 1**)[1]
 - Male patients ages ≥12 years with genetically-confirmed BTHS were randomized (1:1) to subcutaneous (sc) elamipretide 40 mg once daily or placebo for 12 weeks (Treatment Period 1) followed by a 4-week washout period, after which patients were crossed over to the alternate treatment for a further 12 weeks (Treatment Period 2)
 - Trial design also included an open-label extension treatment period with elamipretide for up to 168 additional weeks.

Figure 1. TAZPOWER Study Design

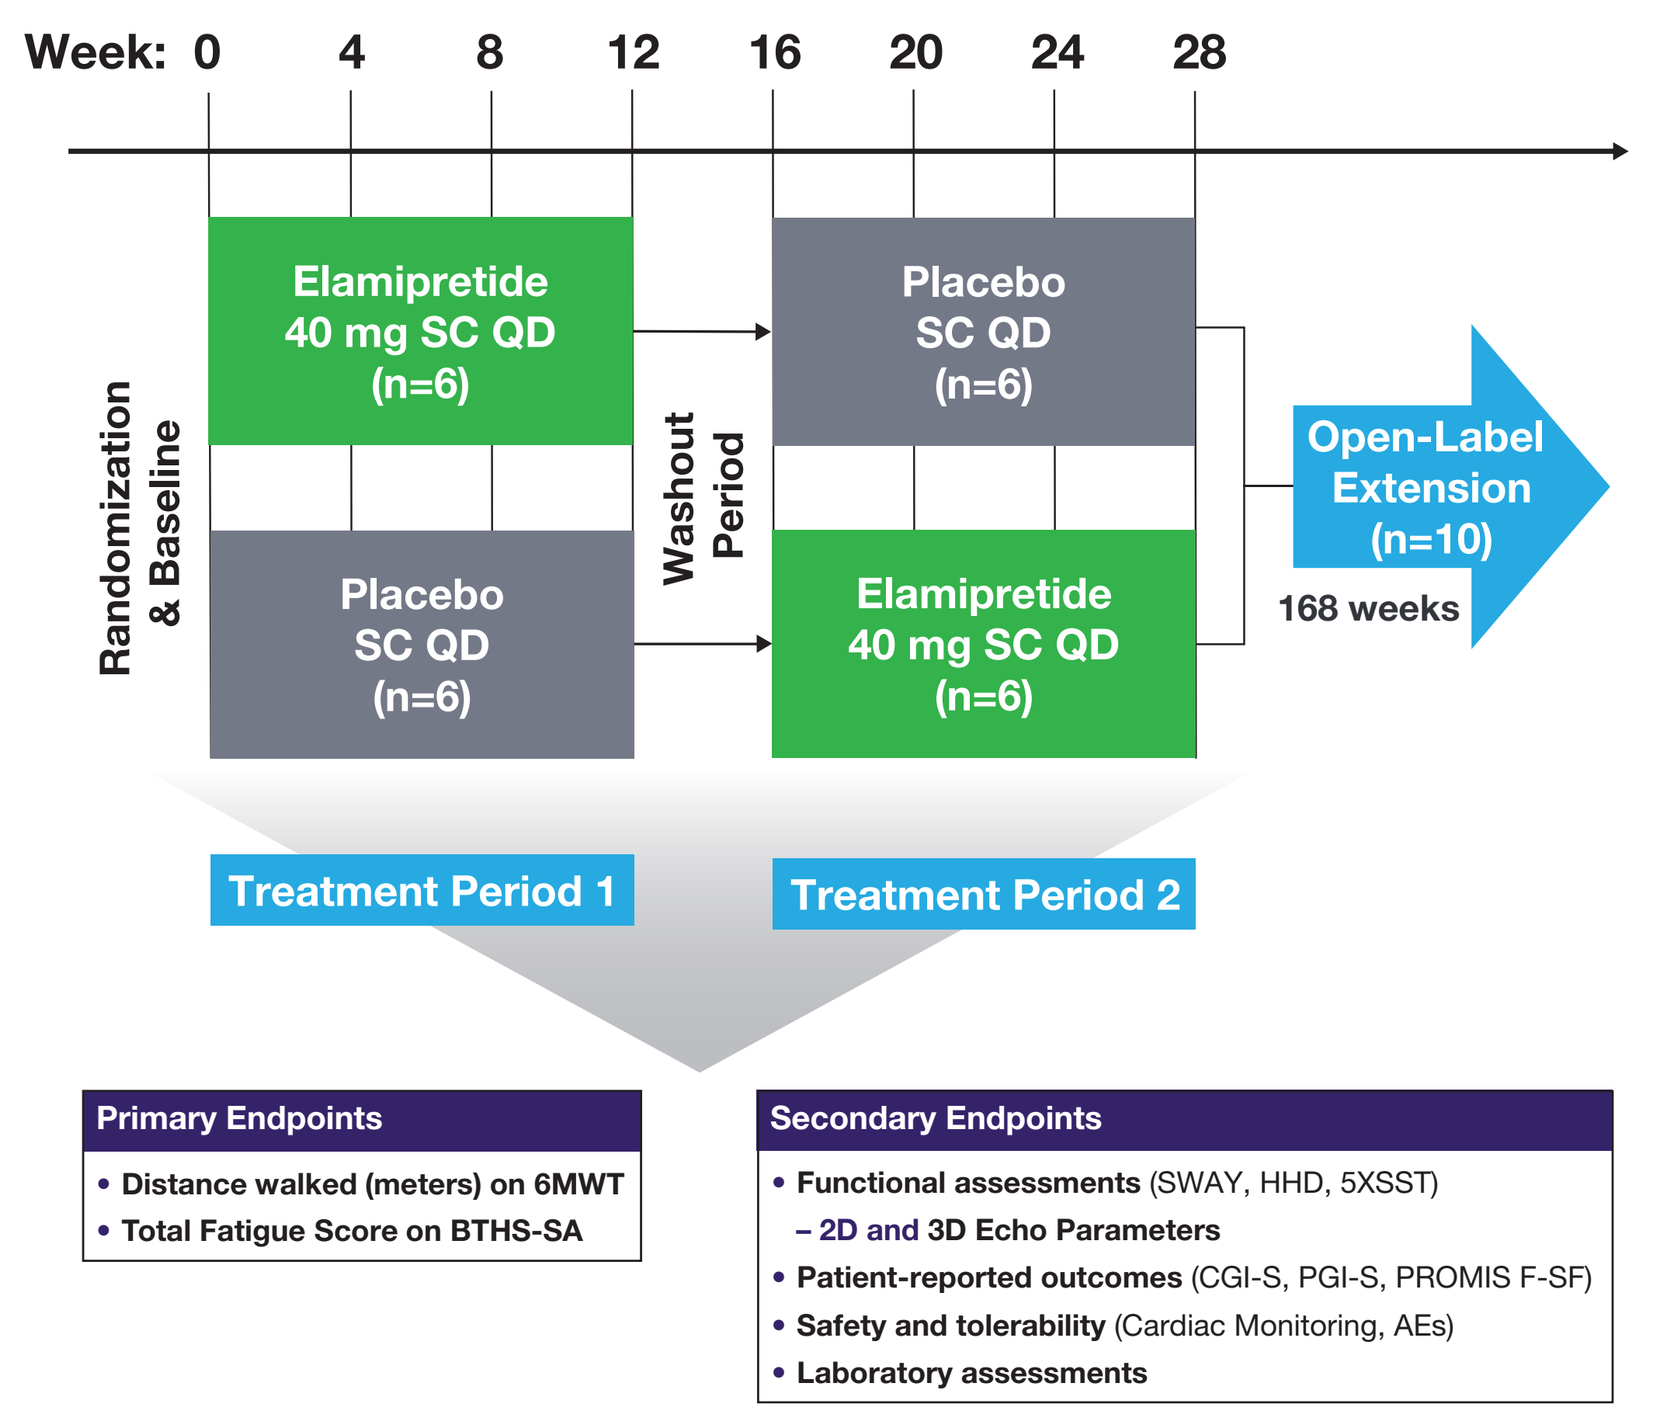
Week: 0 4 8 12 16 20 24 28
Randomization & Baseline
Elamipretide 40 mg SC QD (n=6)
Placebo SC QD (n=6)
Washout Period
Placebo SC QD (n=6)
Elamipretide 40 mg SC QD (n=6)
Open-Label Extension (n=10)
168 weeks
Treatment Period 1
Treatment Period 2
Primary Endpoints
• Distance walked (meters) on 6MWT
• Total Fatigue Score on BTHS-SA
Secondary Endpoints
• Functional assessments (SWAY, HHD, 5XSST)
– 2D and 3D Echo Parameters
• Patient-reported outcomes (CGI-S, PGI-S, PROMIS F-SF)
• Safety and tolerability (Cardiac Monitoring, AEs)
• Laboratory assessments

AEs=Adverse Events; BTHS-SA=BarTH Syndrome Symptom Assessment; CGI-S=Clinician Global Impression of Symptom Severity and Change Scale; 5XSST=Five Times Sit-to-Stand Test; HHD=Hand held Dynamometry; PGI-S=Patient Global Impression of Symptom Severity and Change Scale; PROMIS F-SF=Patient-Reported Outcomes Measurement Information System Fatigue-Short Form; 6MWT=6-Minute Walk Test;SWAY=SWAY Application Balance Assessment.

Metabolomic Analyses

- Patients provided blood and first morning void urine samples on four occasions: (1) pre-drug baseline Treatment Period 1, (2) end of Treatment Period 1, (3) pre-drug baseline Treatment Period 2, and (4) end of Treatment Period 2
- A broad spectrum of plasma and urine metabolites were analysed by liquid chromatography-electrospray ionization tandem mass spectrometry per established protocols at baseline and after 12 weeks of treatment with elamipretide or placebo
- Analysis focused on metabolites previously seen to be altered in BTHS, including acylcarnitines, β-hydroxybutyrate, 3-methylglutaconic acid, and selected circulating amino acids

Statistical Analyses

- Values for urine specimens were normalized to urinary creatinine
- The change (Δ) value for a given metabolite was calculated as the 12-week change from baseline in metabolite concentration with elamipretide treatment minus the 12-week change from baseline with placebo treatment
- The effects of each arm were compared to each other using a paired Student's *t*-test
- Multiple related metabolites were analyzed using a two-way ANOVA, with main factors of metabolite and elamipretide effects, as well as analysis for any interaction among main factors
- Data are presented as means ± standard error
- Significance was noted using an alpha level of $p<0.05$

RESULTS

Patients

- A total of 12 patients were included in TAZPOWER, with 11 patients included in the current analysis (metabolomic data for one patient with diabetes were excluded from the final exploratory analyses, so the complete dataset sample number per metabolite=44)
- Patients were ambulatory but with impaired 6-Minute Walk Test (mean=395.5 meters; range=313–495 meters)
- Vital signs, including ECG parameters, were within normal limits
- Most commonly reported comorbid medical conditions were cardiomyopathy (67%), neutropenia (58%), and hypotonia (50%)

Acylcarnitines

- Acylcarnitines were elevated by 15% in these patients
- Elamipretide treatment significantly reduced levels of plasma acylcarnitines ($p<0.007$) (**Figure 2**)

Figure 2. Change (Δ) in Medium Chain Plasma Acylcarnitines After 12 Weeks Treatment with Placebo or Elamipretide

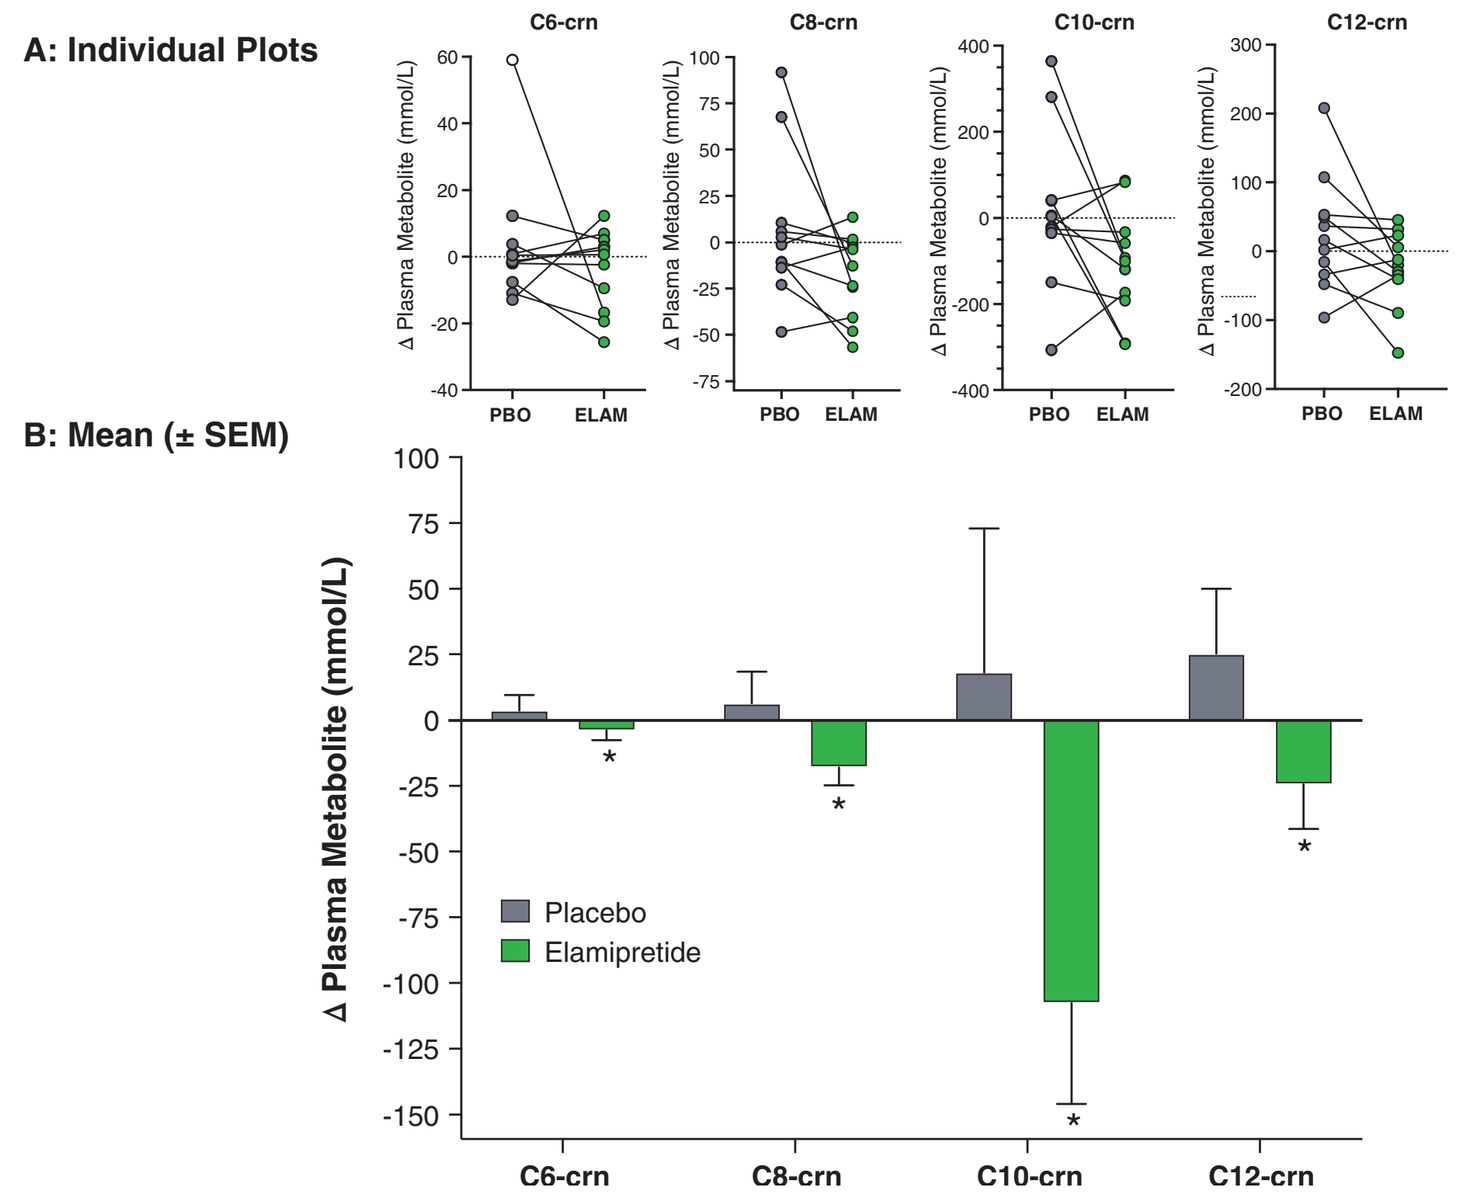
A: Individual Plots
C6-crn
C8-crn
C10-crn
C12-crn
Δ Plasma Metabolite (mmol/L)
PBO
ELAM
B: Mean (± SEM)
Δ Plasma Metabolite (mmol/L)
Placebo
Elamipretide
C6-crn
C8-crn
C10-crn
C12-crn

ELAM=elamipretide; PBO=placebo.
n=11 per group.
*Elamipretide effect ($p=0.007$; ANOVA) for all medium chain (C6-C12) acylcarnitines combined.

- There was a statistically significant reduction in urinary acylcarnitines (C6-C8) with elamipretide compared with placebo ($p=0.0499$ for elamipretide effect) (**Figure 3**)

Figure 3. Mean (± SEM) Change (Δ) in Urinary Acylcarnitines After 12 Weeks Treatment with Placebo or Elamipretide

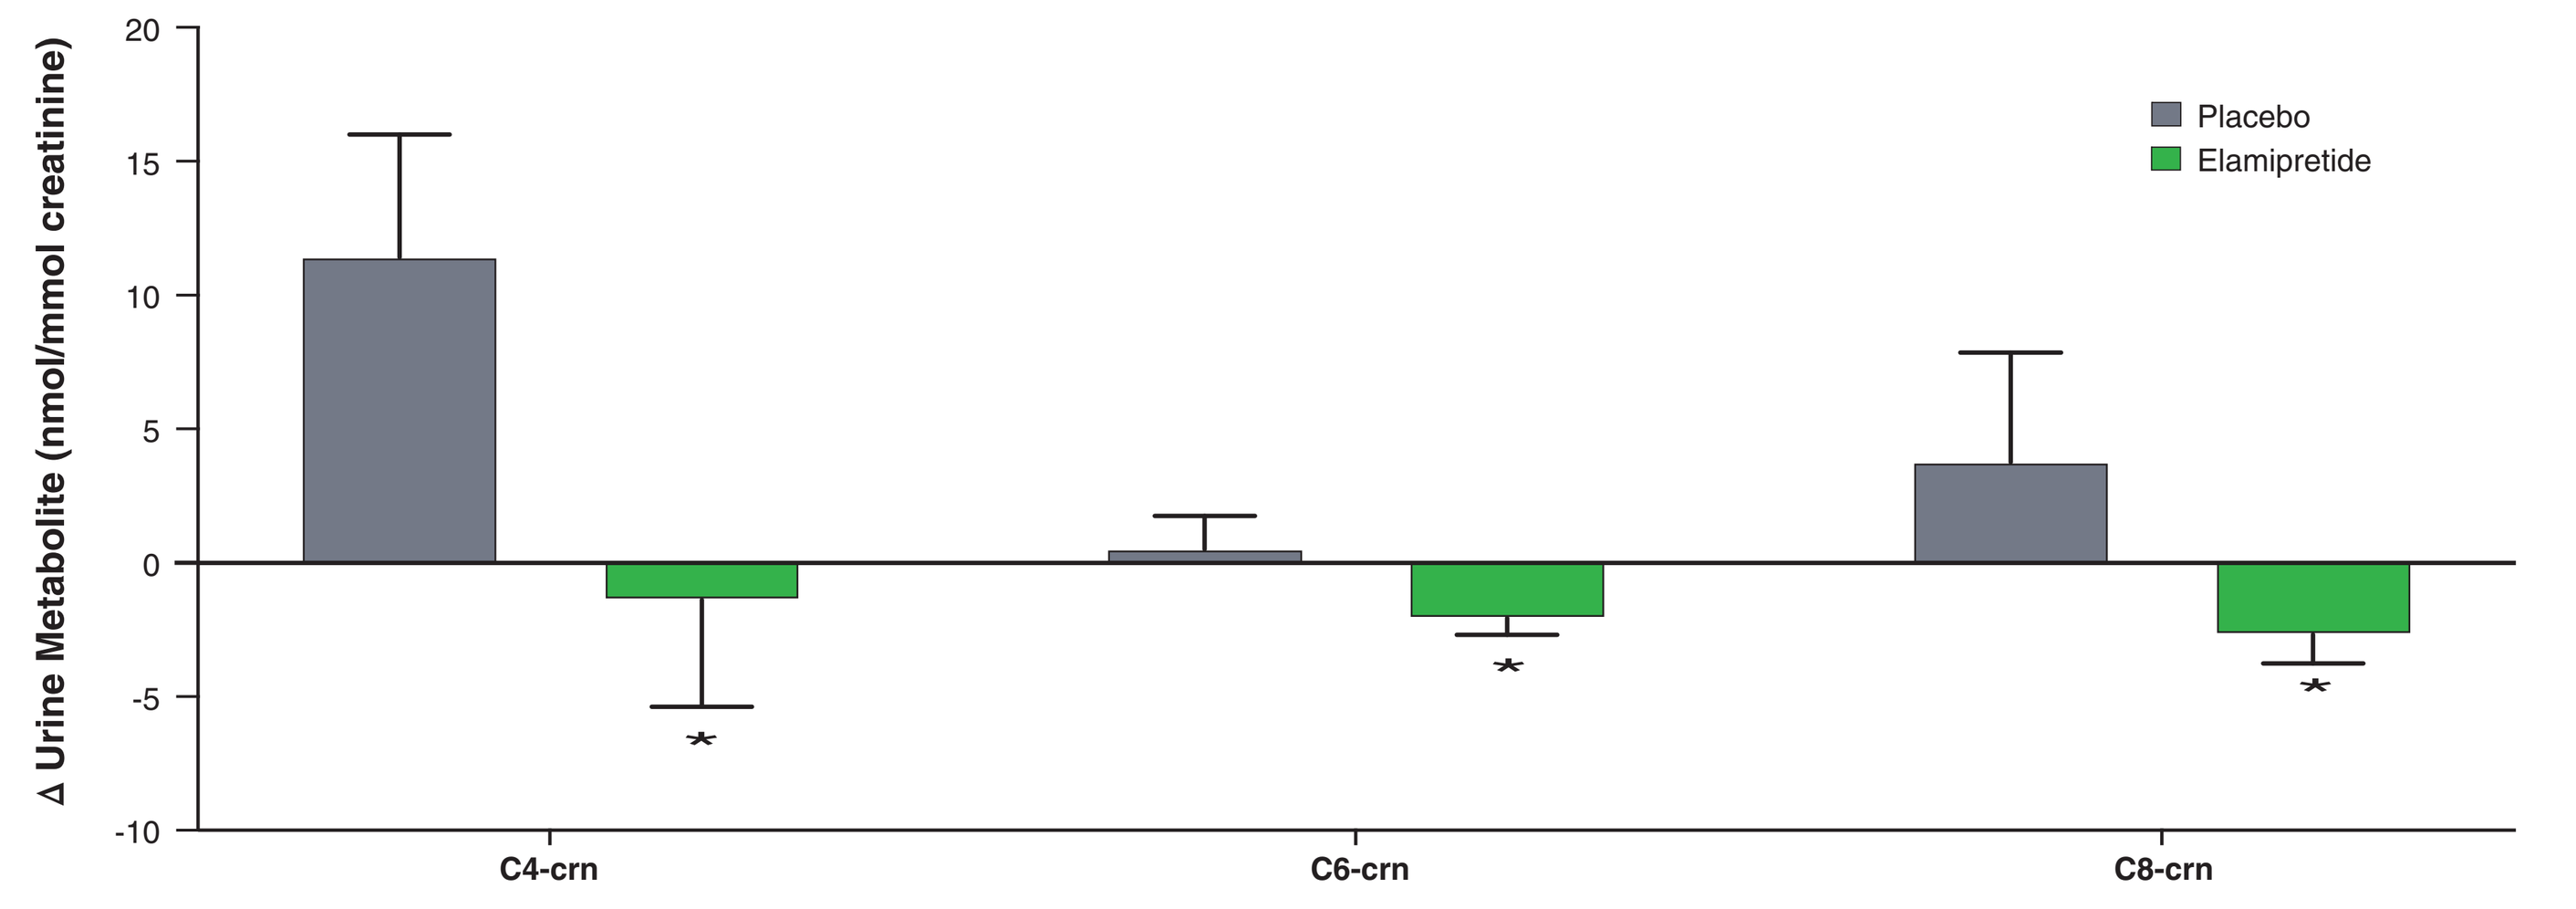
Δ Urine Metabolite (nmol/mmol creatinine)
Placebo
Elamipretide
C4-crn
C6-crn
C8-crn

n=11 per group.
*Elamipretide effect ($p=0.0499$; ANOVA) for C6 and C8 acylcarnitines combined.

Acetylcarnitine

- There was a trend for decreased acetylcarnitine levels in both plasma and urine (normalized to creatinine) following elamipretide treatment, but this decrease did not reach statistical significance ($p=0.13$ for elamipretide effect) (**Figure 4**)

Figure 4. Mean (± SEM) Change (Δ) in Plasma and Urinary Acetylcarnitine (C2-crn) After 12 Weeks Treatment with Placebo or Elamipretide

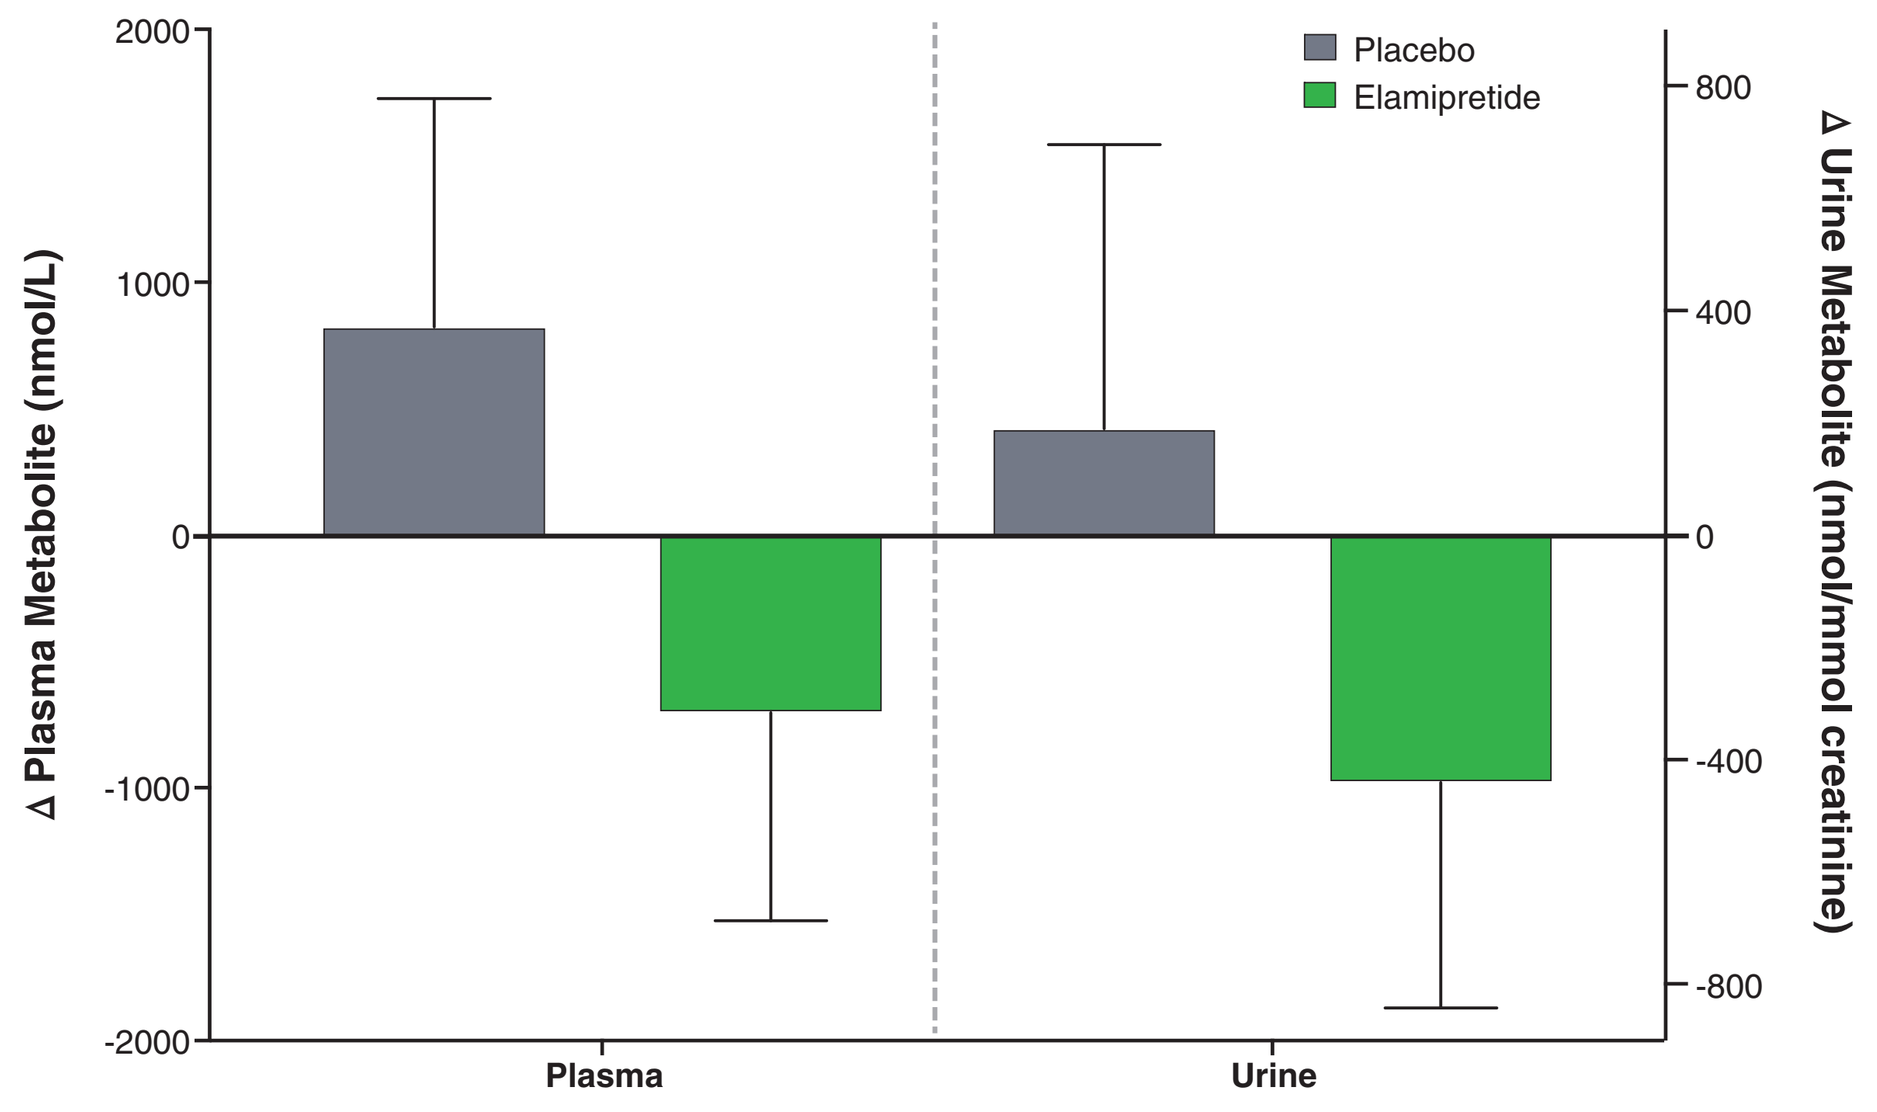
Δ Plasma Metabolite (nmol/L)
Δ Urine Metabolite (nmol/mmol creatinine)
Placebo
Elamipretide
Plasma
Urine

n=11 per group.
Urinary acetylcarnitine concentrations are adjusted for urinary creatinine concentration.

3-Methylglutaconate

- There was a strong trend for decreased 3-methylglutaconate concentration in both plasma and urine, but this did not reach statistical significance compared with placebo ($p=0.084$ for elamipretide effect) (**Figure 5**)

Figure 5. Mean (± SEM) Change (Δ) in Plasma and Urinary 3-Methylglutaconate After 12 Weeks Treatment with Placebo or Elamipretide

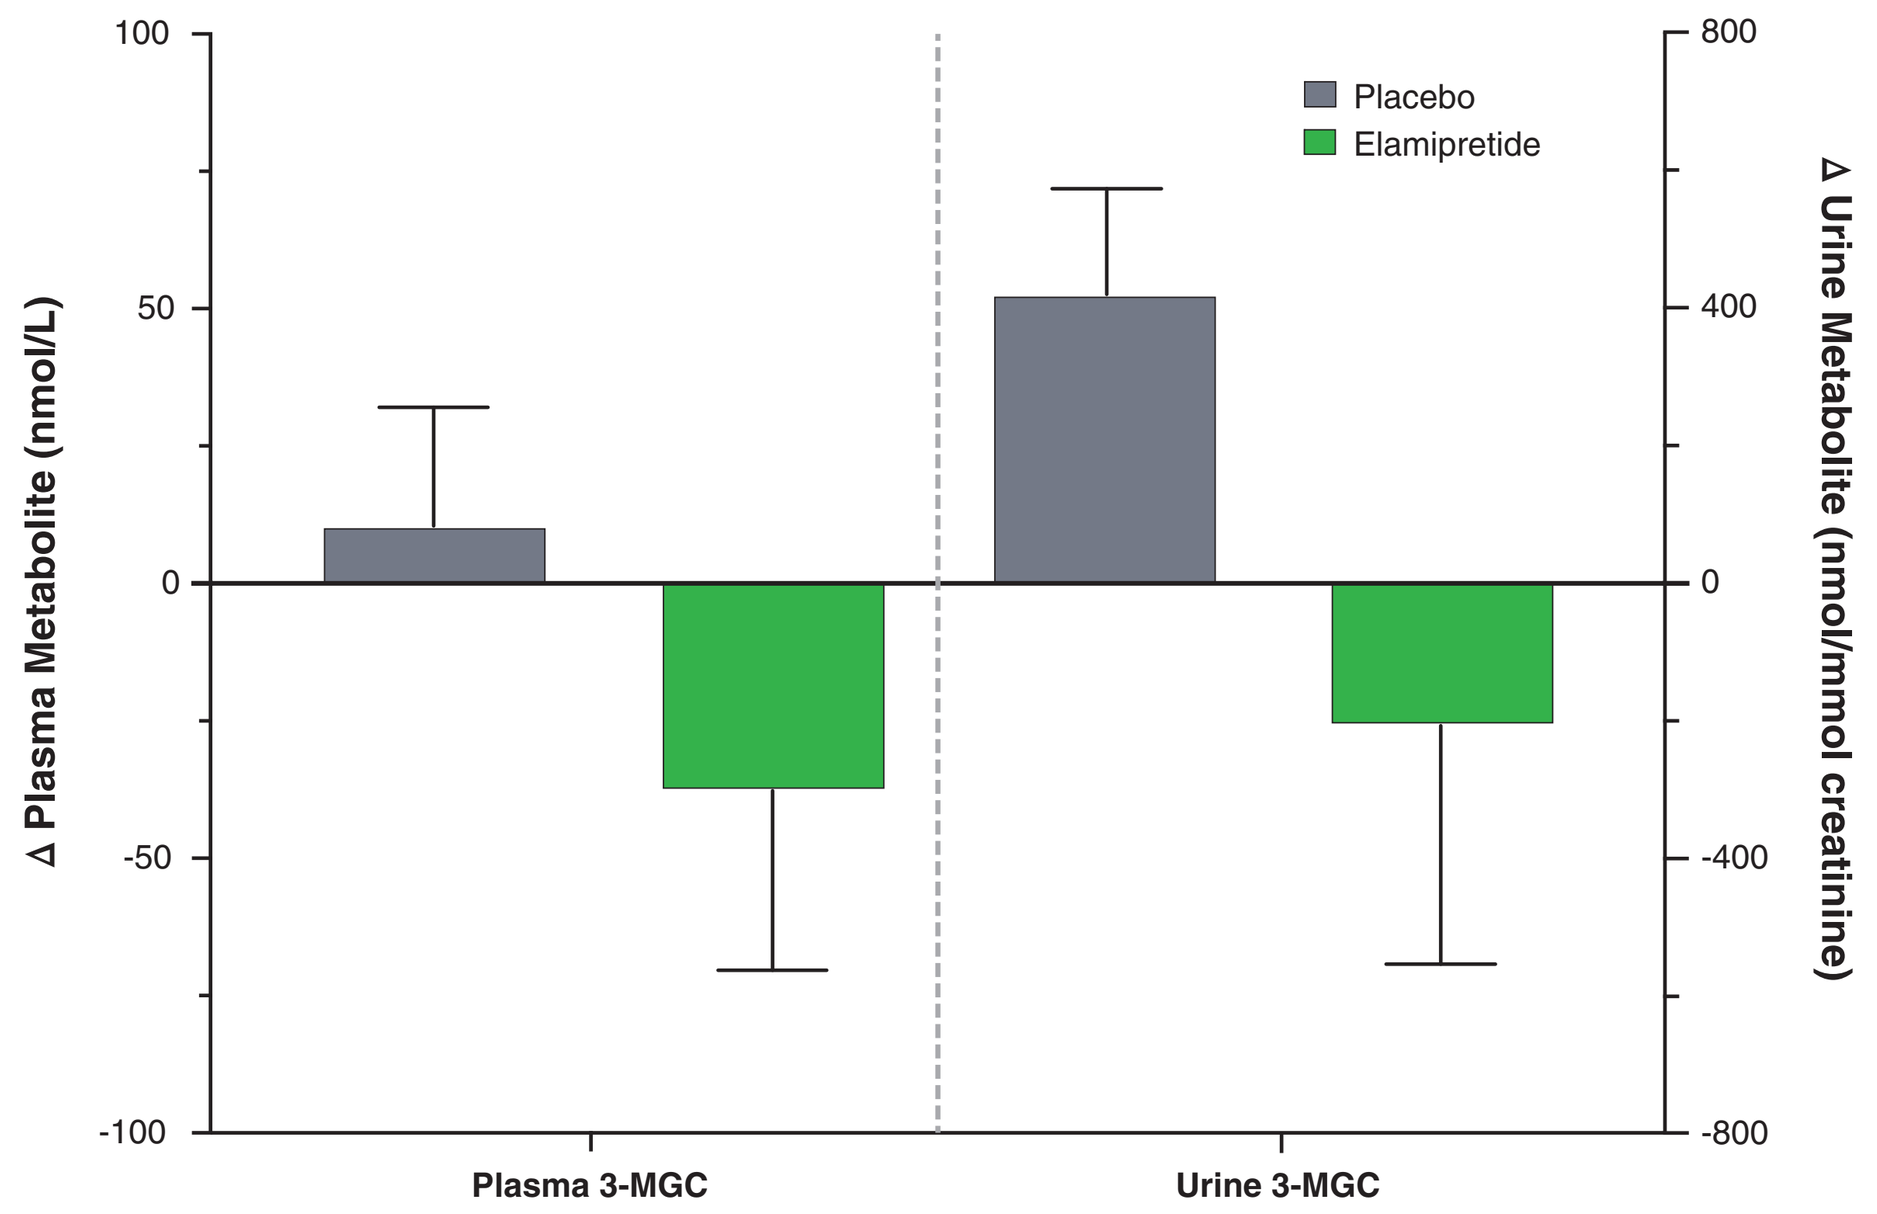
Δ Plasma Metabolite (nmol/L)
Δ Urine Metabolite (nmol/mmol creatinine)
Placebo
Elamipretide
Plasma 3-MGC
Urine 3-MGC

3-MGC=3-Methylglutaconate.
n=11 per group.
Urinary 3-methylglutaconate concentrations are adjusted for urinary creatinine concentration.

3-Hydroxybutyrate

- There was a strong trend for decreased plasma 3-hydroxybutyrate following elamipretide treatment, although this trend did not reach statistical significance compared with placebo ($p=0.07$ for elamipretide effect) (**Figure 6**)

Figure 6. Individual Changes (Δ) in Plasma 3-Hydroxybutyrate Concentration After 12 Weeks Treatment with Placebo or Elamipretide

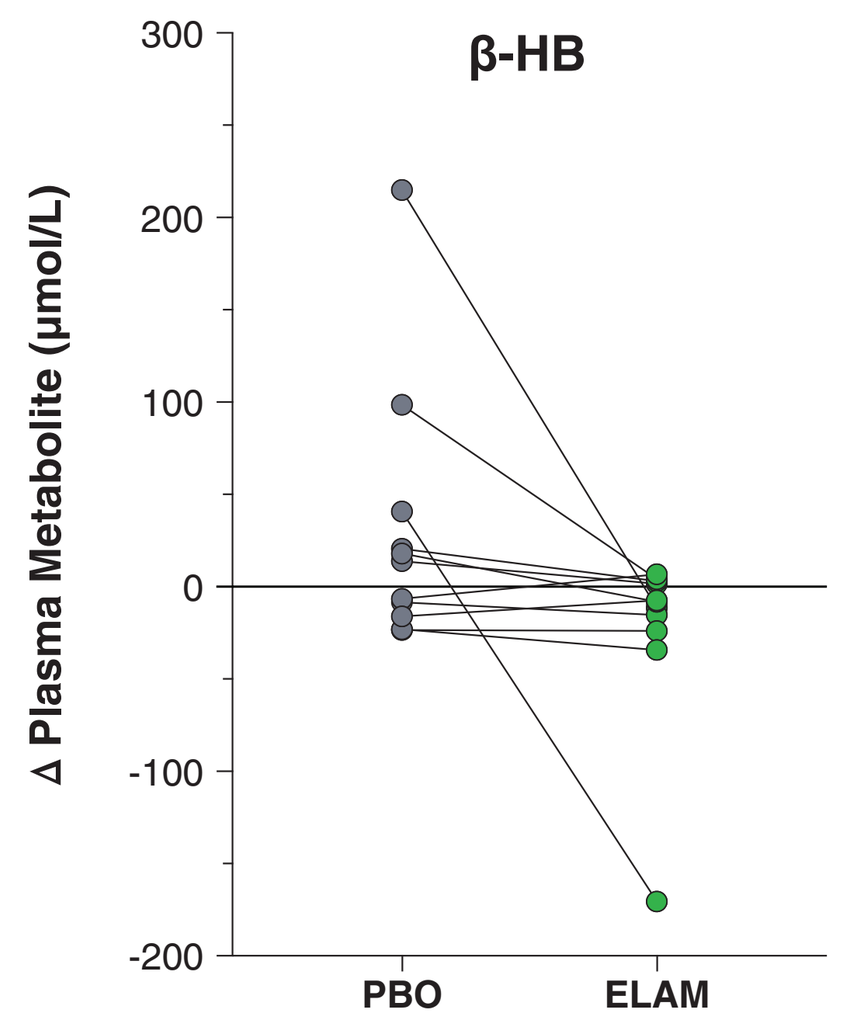
β-HB
Δ Plasma Metabolite (μmol/L)
PBO
ELAM

ELAM=elamipretide; PBO=placebo.
n=11.

Amino Acids

- Baseline plasma concentrations of asparagine were 173 ± 8 mmol/L, a heightened level that corroborates previous findings of elevated plasma asparagine in BTHS
- Plasma concentrations of proline (125 ± 4 mmol/L), tyrosine (57 ± 3 mmol/L), and arginine (54 ± 4 mmol/L) were within normal ranges previously seen in non-BTHS control patients
- There were no statistically significant changes in the elamipretide arm for any of these four amino acids (**Table 1**)

Table 1. Change in Plasma Amino Acid Concentrations After 12 Weeks Treatment with Elamipretide or Placebo

	Mean (SEM) Change in Concentration (mmol/L) after 12 Weeks vs Baseline		
Amino acid	Placebo	Elamipretide	*p*-value
Asparagine	–32.7 (14.6)	–2.2 (17.5)	0.16
Proline	–5.1 (6.9)	0.35 (6.7)	0.60
Tyrosine	0.18 (4.3)	–1.3 (3.3)	0.81
Arginine	–3.7 (7.4)	–6.0 (5.5)	0.81

SEM=standard error of the mean.

Taurine

- Compared with placebo, circulating taurine increased significantly in the elamipretide arm of the study ($p=0.022$ for elamipretide effect, paired *t*-test) (**Figure 7**)

Figure 7. Individual Changes (Δ) in Plasma Taurine Concentration After 12 Weeks Treatment with Placebo or Elamipretide

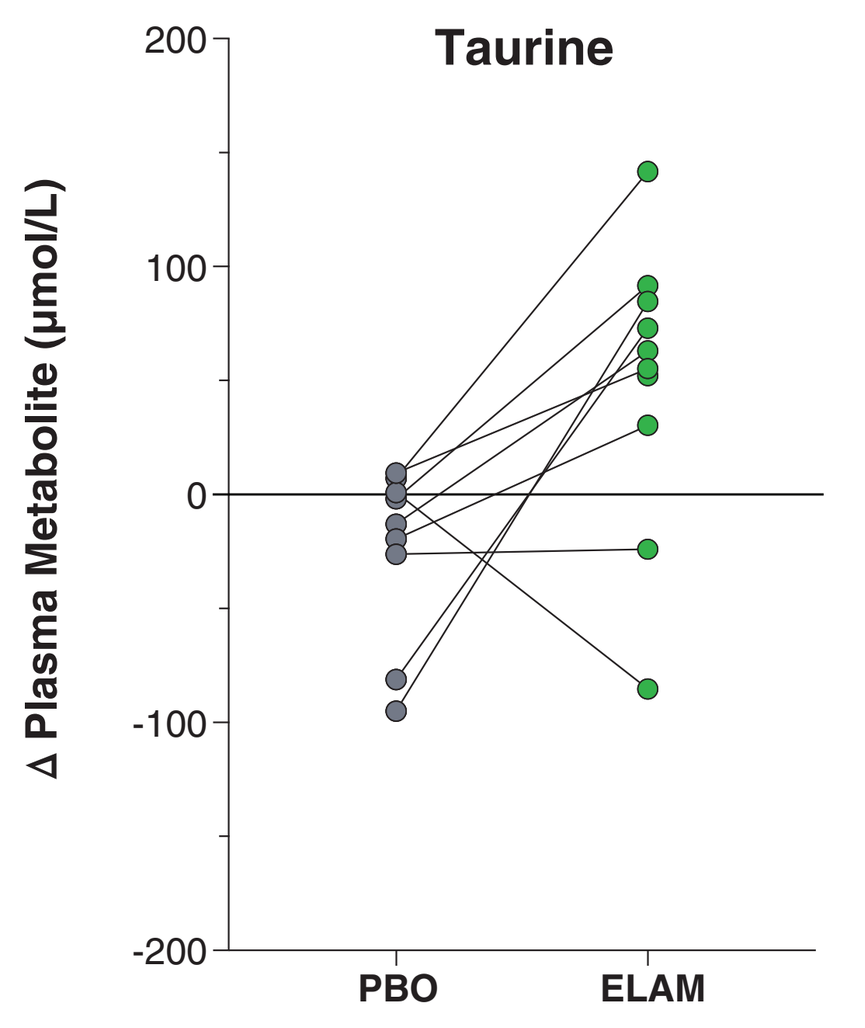
Taurine
Δ Plasma Metabolite (μmol/L)
PBO
ELAM

ELAM=elamipretide; PBO=placebo.
n=10.

CONCLUSIONS

- Alterations in several metabolic biomarkers were observed in TAZPOWER, and are reflective of impaired mitochondrial function in BTHS
- As evidenced by reduced plasma and urinary content of acylcarnitines and trends for lowered ketone body 3-hydroxybutyrate, fat metabolism in BTHS appears to be improved with 12 weeks of elamipretide treatment
- After 12 weeks of treatment, elamipretide had no discernible effect on four amino acids previously characterized as having abnormal concentrations in patents with BTHS
- Elamipretide caused a significant rise in plasma taurine concentrations, a protective amino acid which has been observed to be decreased in patients with BTHS
- These data further highlight the potential of elamipretide to treat skeletal muscle wasting and cardiomyopathies and are consistent with the improved mitochondrial function that may precede functional benefits with a longer duration of therapy with elamipretide in patients with BTHS

REFERENCE

1. Thompson RW, Hornby B, Manuel R, Bradley E, Laux J, Carr J, Vernon HJ. A phase 2/3 randomized clinical trial followed by an open-label extension to evaluate the effectiveness of elamipretide in Barth syndrome, a genetic disorder of mitochondrial cardiolipin metabolism. *Genet Med.* 2020 Oct 20. doi: 10.1038/s41436-020-01006-8. Epub ahead of print. PMID: 33077895.

Disclosures:
Funding Source: TAZPOWER Trial and metabolomic analyses funded by Stealth BioTherapeutics, Newton, MA
Acknowledgements: Peter J Oates, PhD, Oates Biomedical Consulting, provided data analysis and support, and James A. Shiffer RPh, Write On Time Medical Communications, LLC provided medical writing assistance