Elamipretide Improves Skeletal Muscle Function in Elderly Subjects: Results from a Randomized, Double-Blind, Placebo-Controlled, Single IV-Dose Study

UNIVERSITY of WASHINGTON

Authors: Conley KE, PhD,[1] Liu Z, PhD,[1] Ali AS,[1] Amory JK, MD,[1] Robertson HL, MD,[1] Goss C,[1] Shankland EG, PhD,[1] Marcinek DM, PhD,[1] and Roshanravan B, MD[1]

[1]Department of Radiology, University of Washington Medical Center, Seattle, WA 98195, USA

Stealth BIOTHERAPEUTICS

ABSTRACT

Introduction: Mitochondrial dysfunction leads to reduced supply of ATP and may lead to overproduction of toxic levels of intracellular reactive oxygen species (ROS). Excess ROS results in oxidative damage to mitochondrial inner membrane structures and to cardiolipin, which stabilizes the inner membrane. Oxidative damage to mitochondria leads to sarcopenia and exercise intolerance. Mitochondrial dysfunction has been documented in heart failure (HF) patients, and exercise intolerance is a hallmark symptom of the disease. Currently available treatments have not demonstrated the ability to improve skeletal muscle function in HF patients. The results of this trial, therefore, can have direct clinical implications to HF patients.

Hypothesis: Elamipretide (formerly referred to as Bendavia, SS-31 and MTP-131 [ELAM]) is a mitochondria-targeting peptide that readily penetrates cell membranes and outer mitochondrial membranes to localize on the inner membranes of mitochondria, where it associates with cardiolipin. In doing so, it improves the integrity and efficiency of the electron transport chain (ETC), thereby improving ATP synthesis and reducing the production of ROS. ELAM effectively improved muscle function and biomarker levels in multiple animal models of dysfunction (specifically, age-related skeletal muscle immobilization, and chronic skeletal muscle dysfunction). We studied the effect of ELAM on skeletal muscle energetics and performance in elderly subjects.

Methods: Elderly subjects (≥60 and ≤85 yrs) with documented mitochondrial dysfunction received a single IV dose of ELAM 0.25 mg/(kg • hr), or placebo, infused at 60 mL/hr for 2 hrs (randomized 1:1). On the infusion day and day 7, maximum mitochondrial ATP synthesis (ATP_{max}), and mitochondrial coupling of ATP synthesis per O_2 uptake (P/O) were measured *in vivo* using phosphorous magnetic resonance spectroscopy (ATP synthesis). Optical spectroscopy determined O_2 uptake. A sustained hand fatigue test determined the effect of increasing ATP_{max} on exercise tolerance because hand-fatigue testing is representative of skeletal muscle measures. Mean changes from baseline were compared between treatment groups using analysis of covariance (baseline as a covariate).

Results: ELAM treatment was evaluated for mitochondrial improvements on infusion day (Visit 2; change from baseline) for ATP_{max}, and 7 days after infusion (Visit 4) for ATP_{max}, P/O, and O_2 uptake. Muscle exercise tolerance was also evaluated 2 hrs after infusion (day 2) and on day 7.

Conclusions: ELAM treatment, in a single infusion, was evaluated for its potential to reverse mitochondrial dysfunction, and O_2 demand, and improve sustained exercise performance in the muscle of elderly subjects. ELAM may potentially target the mitochondrial dysfunction that exists in patients with HF, thereby improving sarcopenia and exercise tolerance. A statistical analysis of the results allowed for a comparison to previous studies in elderly patients that demonstrated a similar 30% increase in ATP_{max} post 6-months of endurance training.[1]

INTRODUCTION

- Heart failure patients are frequently elderly with poor quality of life (QoL)[6-8], exercise intolerance, fatigue, and muscle atrophy
- Given the high energy demands of skeletal muscle, heart, and kidney, mitochondrial dysfunction is postulated to be a contributing factor[3-5]
- Six months of endurance training has been shown to improve bioenergetics in the elderly, but no approved therapies have demonstrated efficacy

Figure 1. Age-related changes in muscle physiology and the relationship between musclemitochondrial capacity/efficiency, aerobic capacity, and walking speed in older adults.[8]

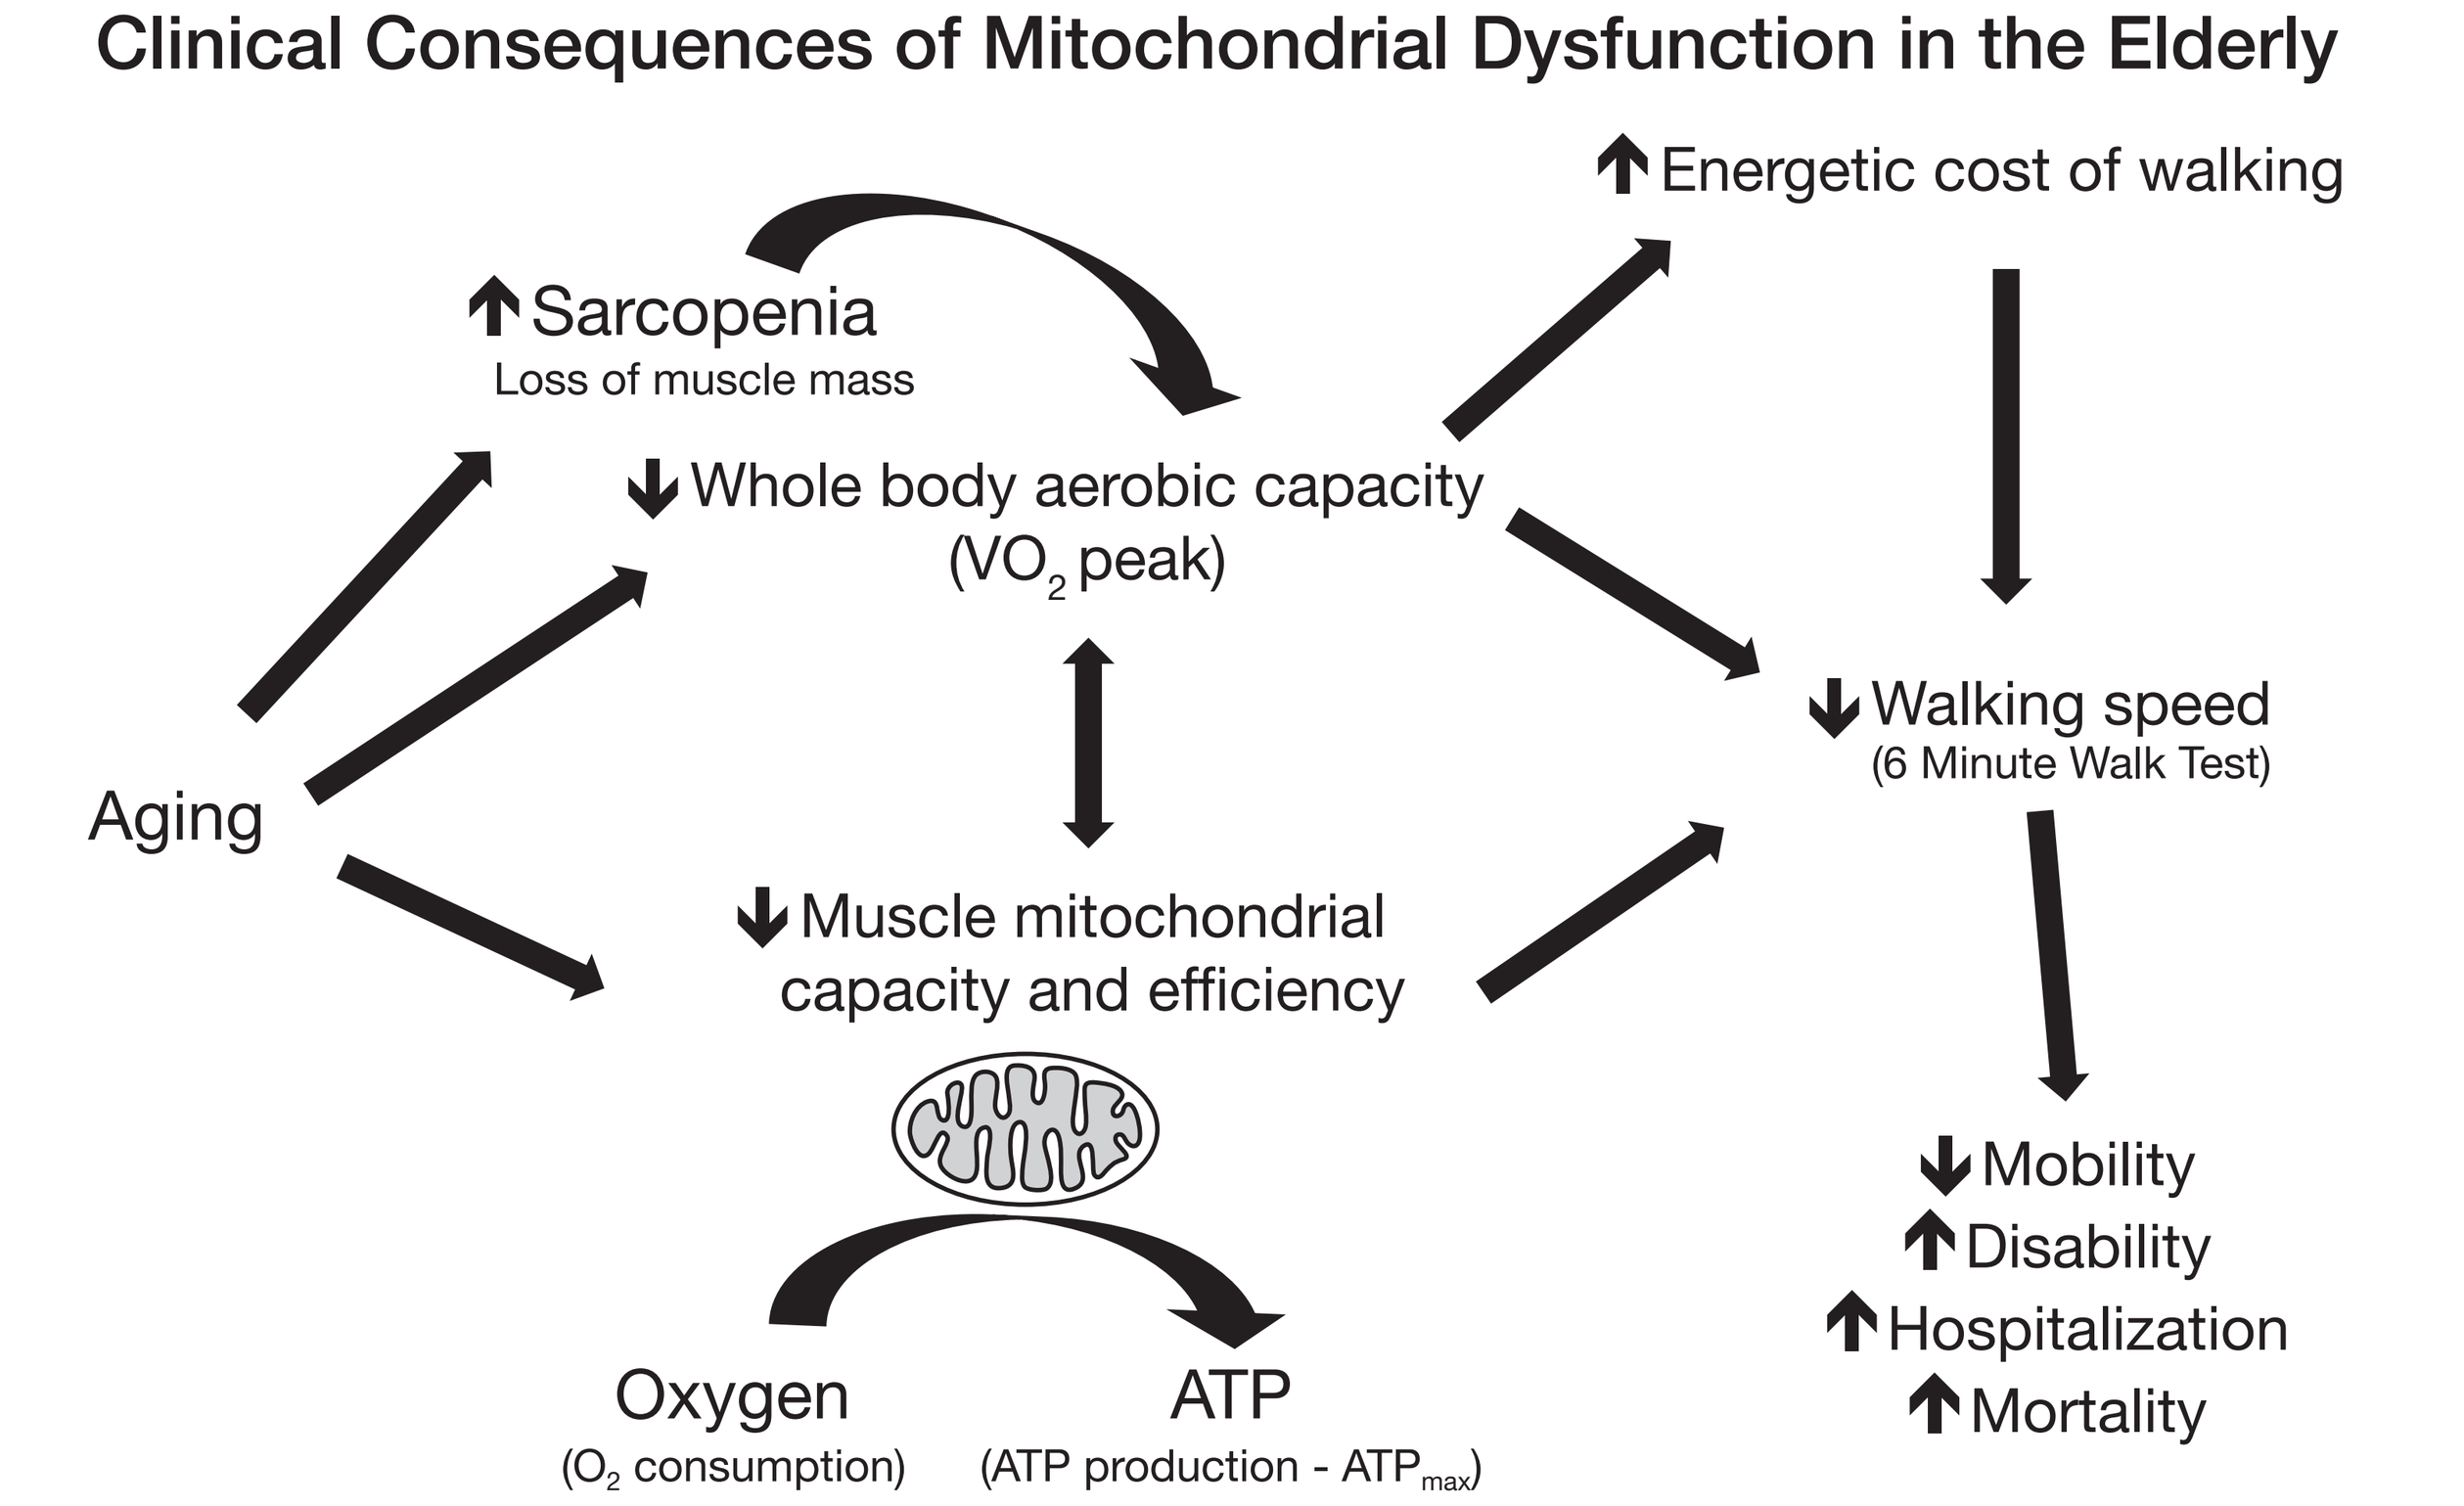
Clinical Consequences of Mitochondrial Dysfunction in the Elderly
↑Energetic cost of walking
↑Sarcopenia
Loss of muscle mass
↓Whole body aerobic capacity (VO_2 peak)
↓Walking speed
(6 Minute Walk Test)
Aging
↓Muscle mitochondrial capacity and efficiency
↓Mobility
↑Disability
↑Hospitalization
↑Mortality
Oxygen
(O_2 consumption)
ATP
(ATP production - ATP_{max})

VO_2 peak = maximal oxygen consumption during maximal dynamic exercise.

INTRODUCTION (cont.)

Interventions

- ELAM selectively associates with cardiolipin, stabilizing ETC supercomplexes and mitochondrial cristae, which are essential for mitochondrial bioenergetics (**Figure 2**)[9,10]
- ELAM reduces mitochondrial ROS production, but is not a ROS scavenger
- Pre-clinical studies have demonstrated improved ATP production and exercise function in aged mice (**Figure 3**)

Figure 2. Beneficial effects of ELAM on mitochondrial function.[11]

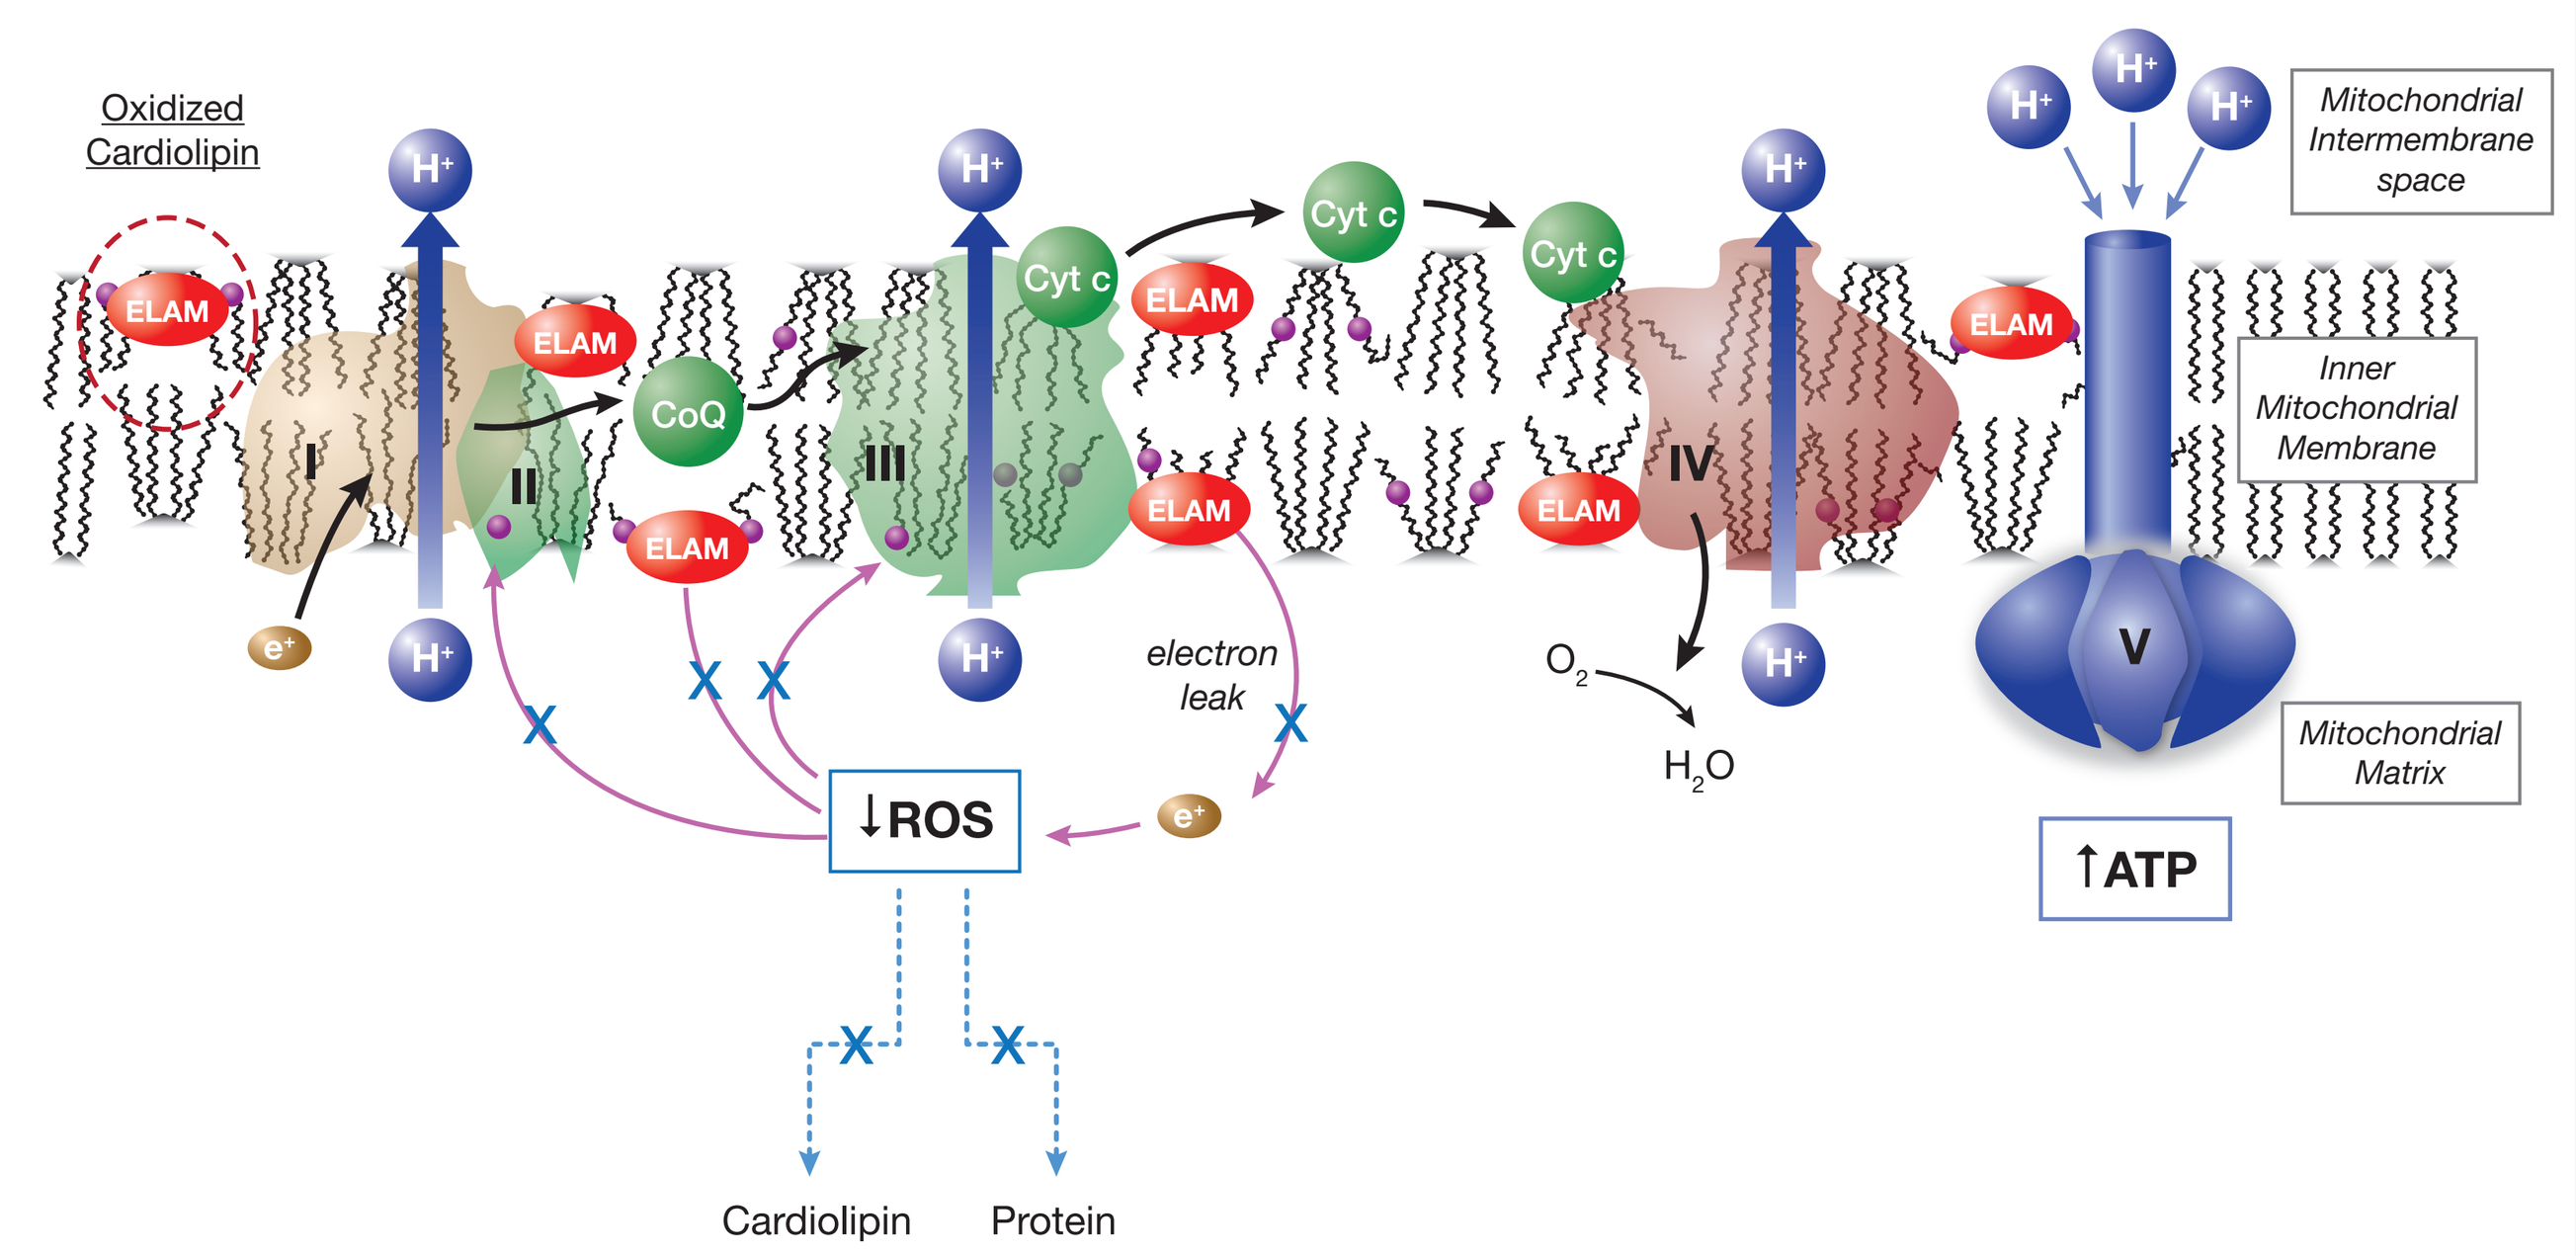
Oxidized Cardiolipin
ELAM
CoQ
Cyt c
I
II
III
IV
V
H^+
e^+
electron leak
O_2
H_2O
↓ROS
↑ATP
Cardiolipin
Protein
Mitochondrial Intermembrane space
Inner Mitochondrial Membrane
Mitochondrial Matrix

Adapted with permission from: Brown DA, et al. *Pharmacol Ther*. 2013 Dec;140(3):258-266.

Figure 3. ELAM improves ATP production and exercise function in mice.[12]

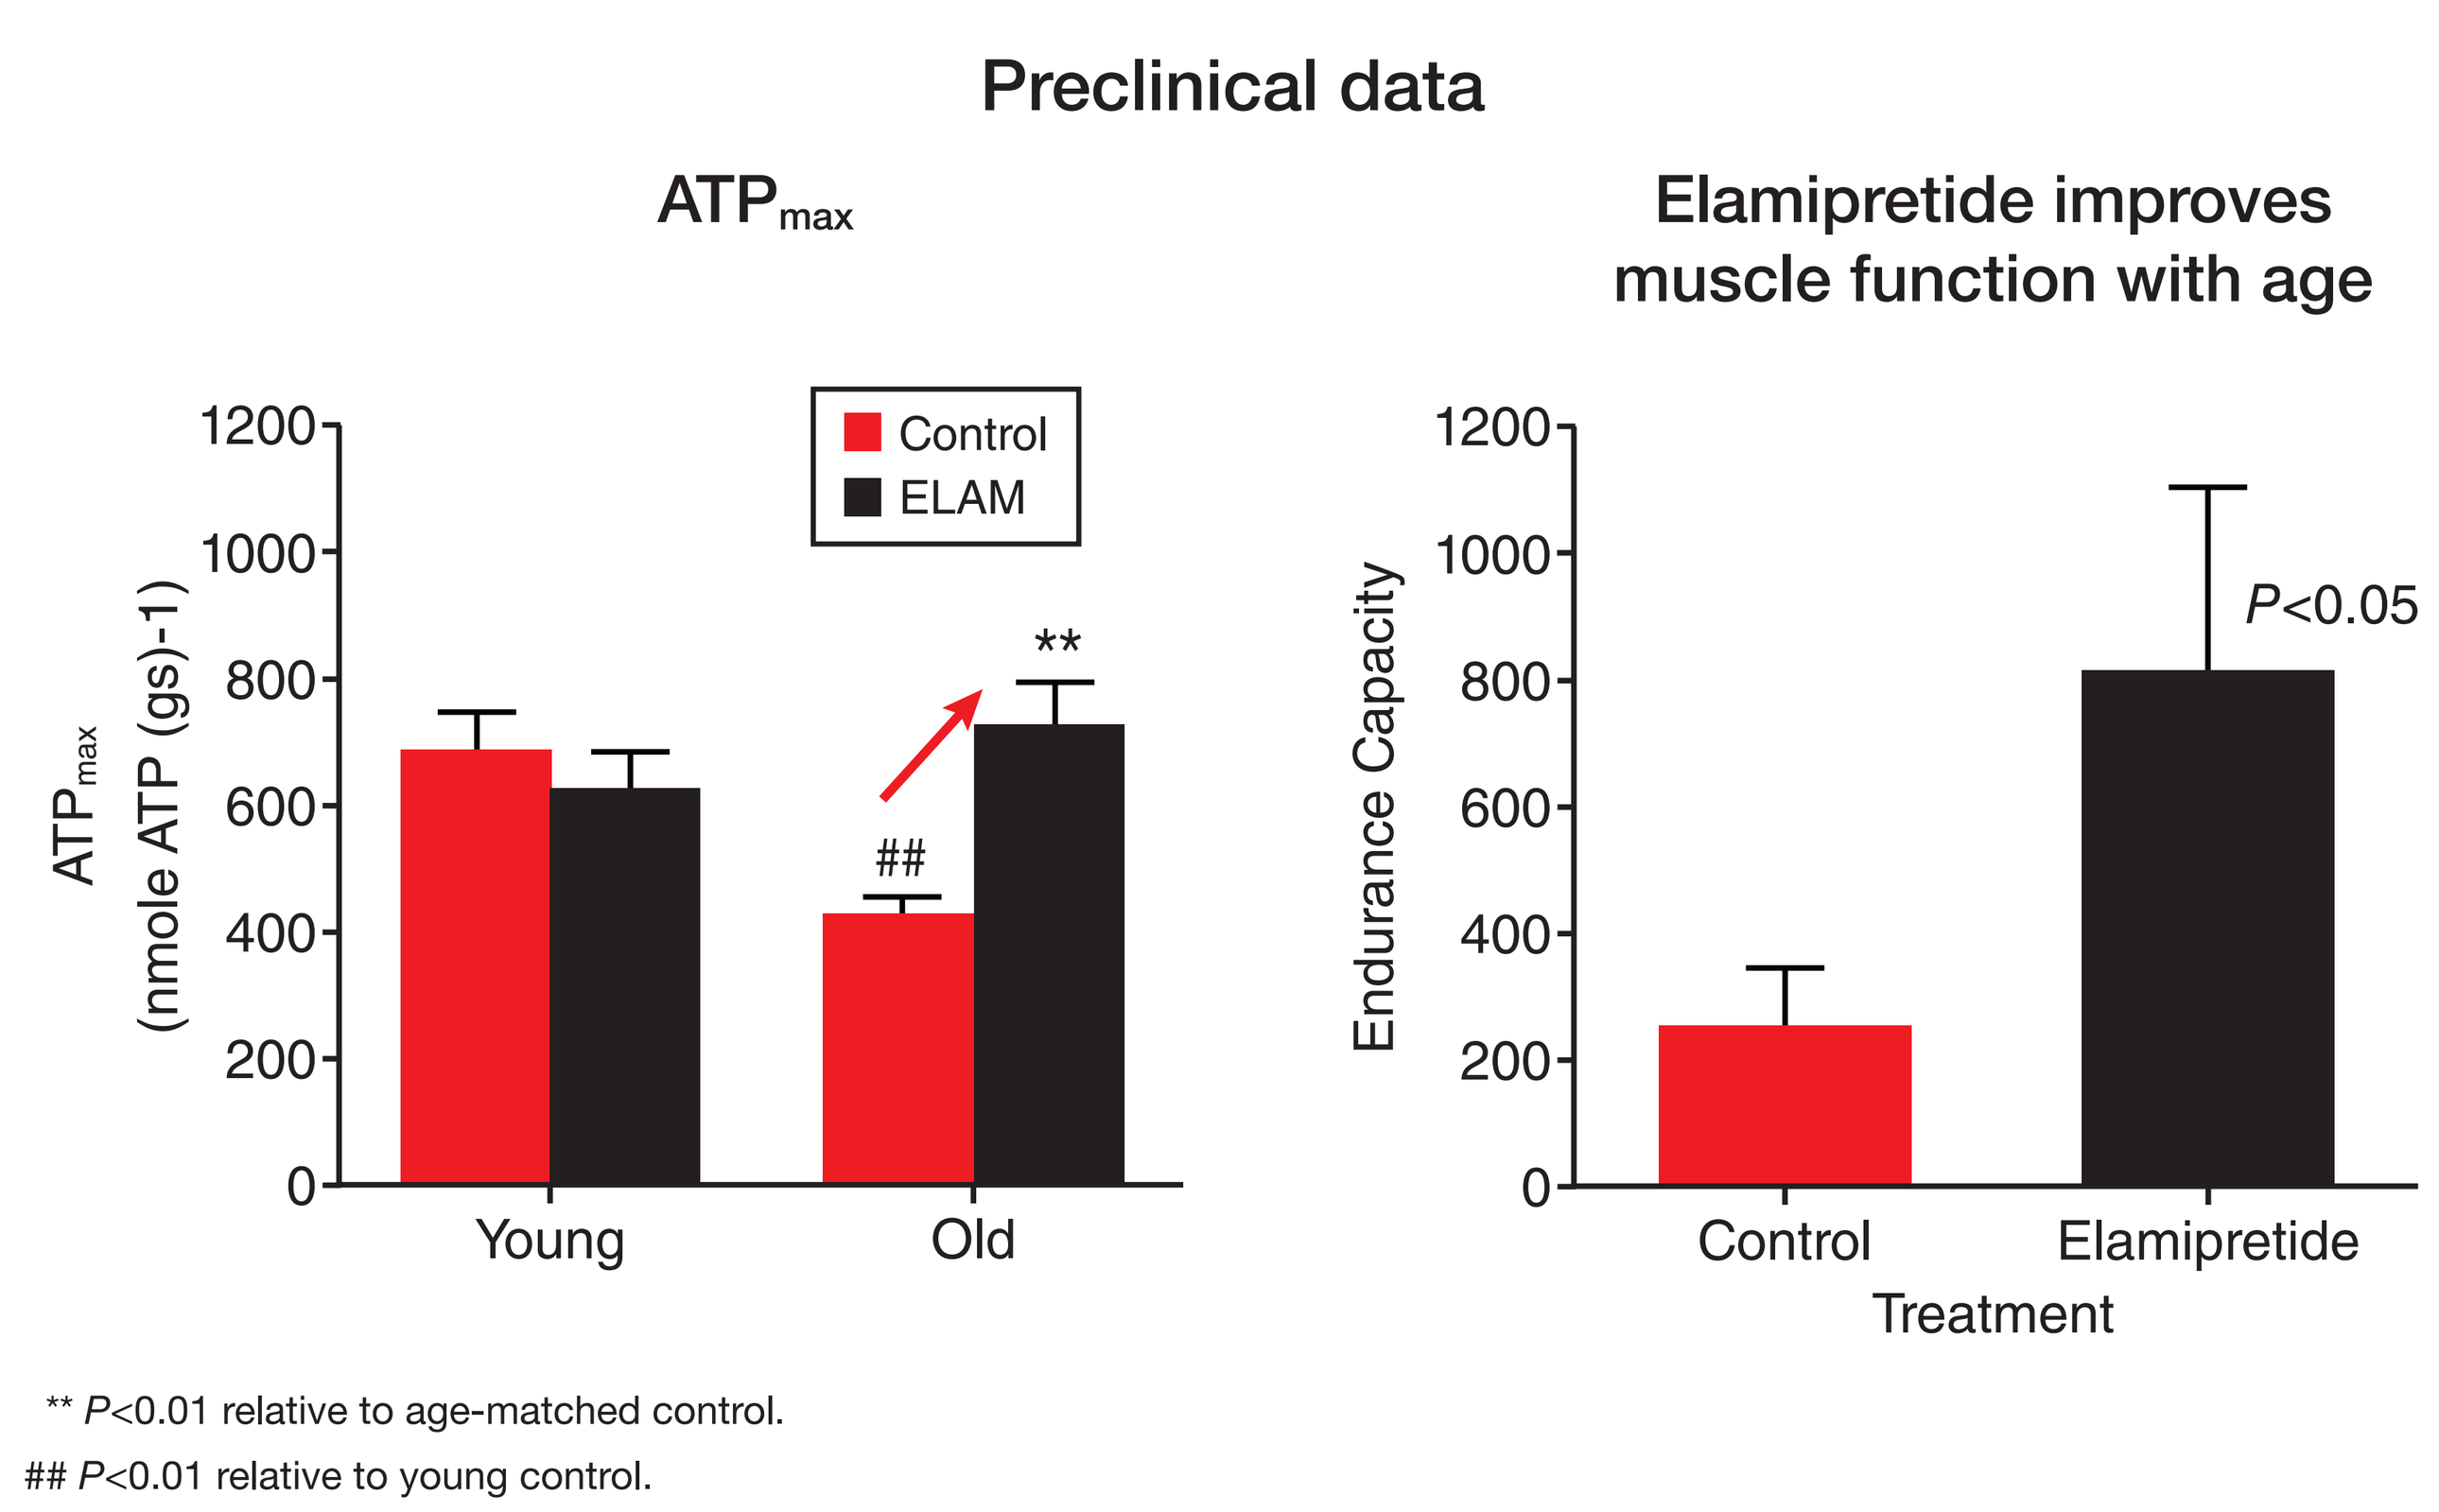
Preclinical data
ATP_{max}
Control
ELAM
ATP_{max} (nmole ATP (gs)-1)
1200
1000
800
600
400
200
0
##
**
Young
Old
Elamipretide improves muscle function with age
Endurance Capacity
P<0.05
Control
Elamipretide
Treatment

** *P*<0.01 relative to age-matched control.
P<0.01 relative to young control.

STUDY OBJECTIVE

A Phase 2 randomized, double-blind, placebo-controlled study sought to evaluate the ability of a single intravenous dose of ELAM to improve skeletal muscle function in elderly subjects

Primary

- Evaluate the effect of ELAM, given as an intravenous (IV) infusion, on hand skeletal muscle energetics and muscle performance as measured by *in vivo* 31 phosphorus-31 (31 P) magnetic resonance spectroscopy (MRS), *in vivo* optical spectra scan (OPS), and a muscle fatigue test in elderly subjects with evidence of mitochondrial dysfunction

Secondary

- Assess the safety and tolerability of a single IV infusion of ELAM in elderly subjects with evidence of skeletal muscle mitochondrial dysfunction

Figure 4. Measuring change in muscle performance.

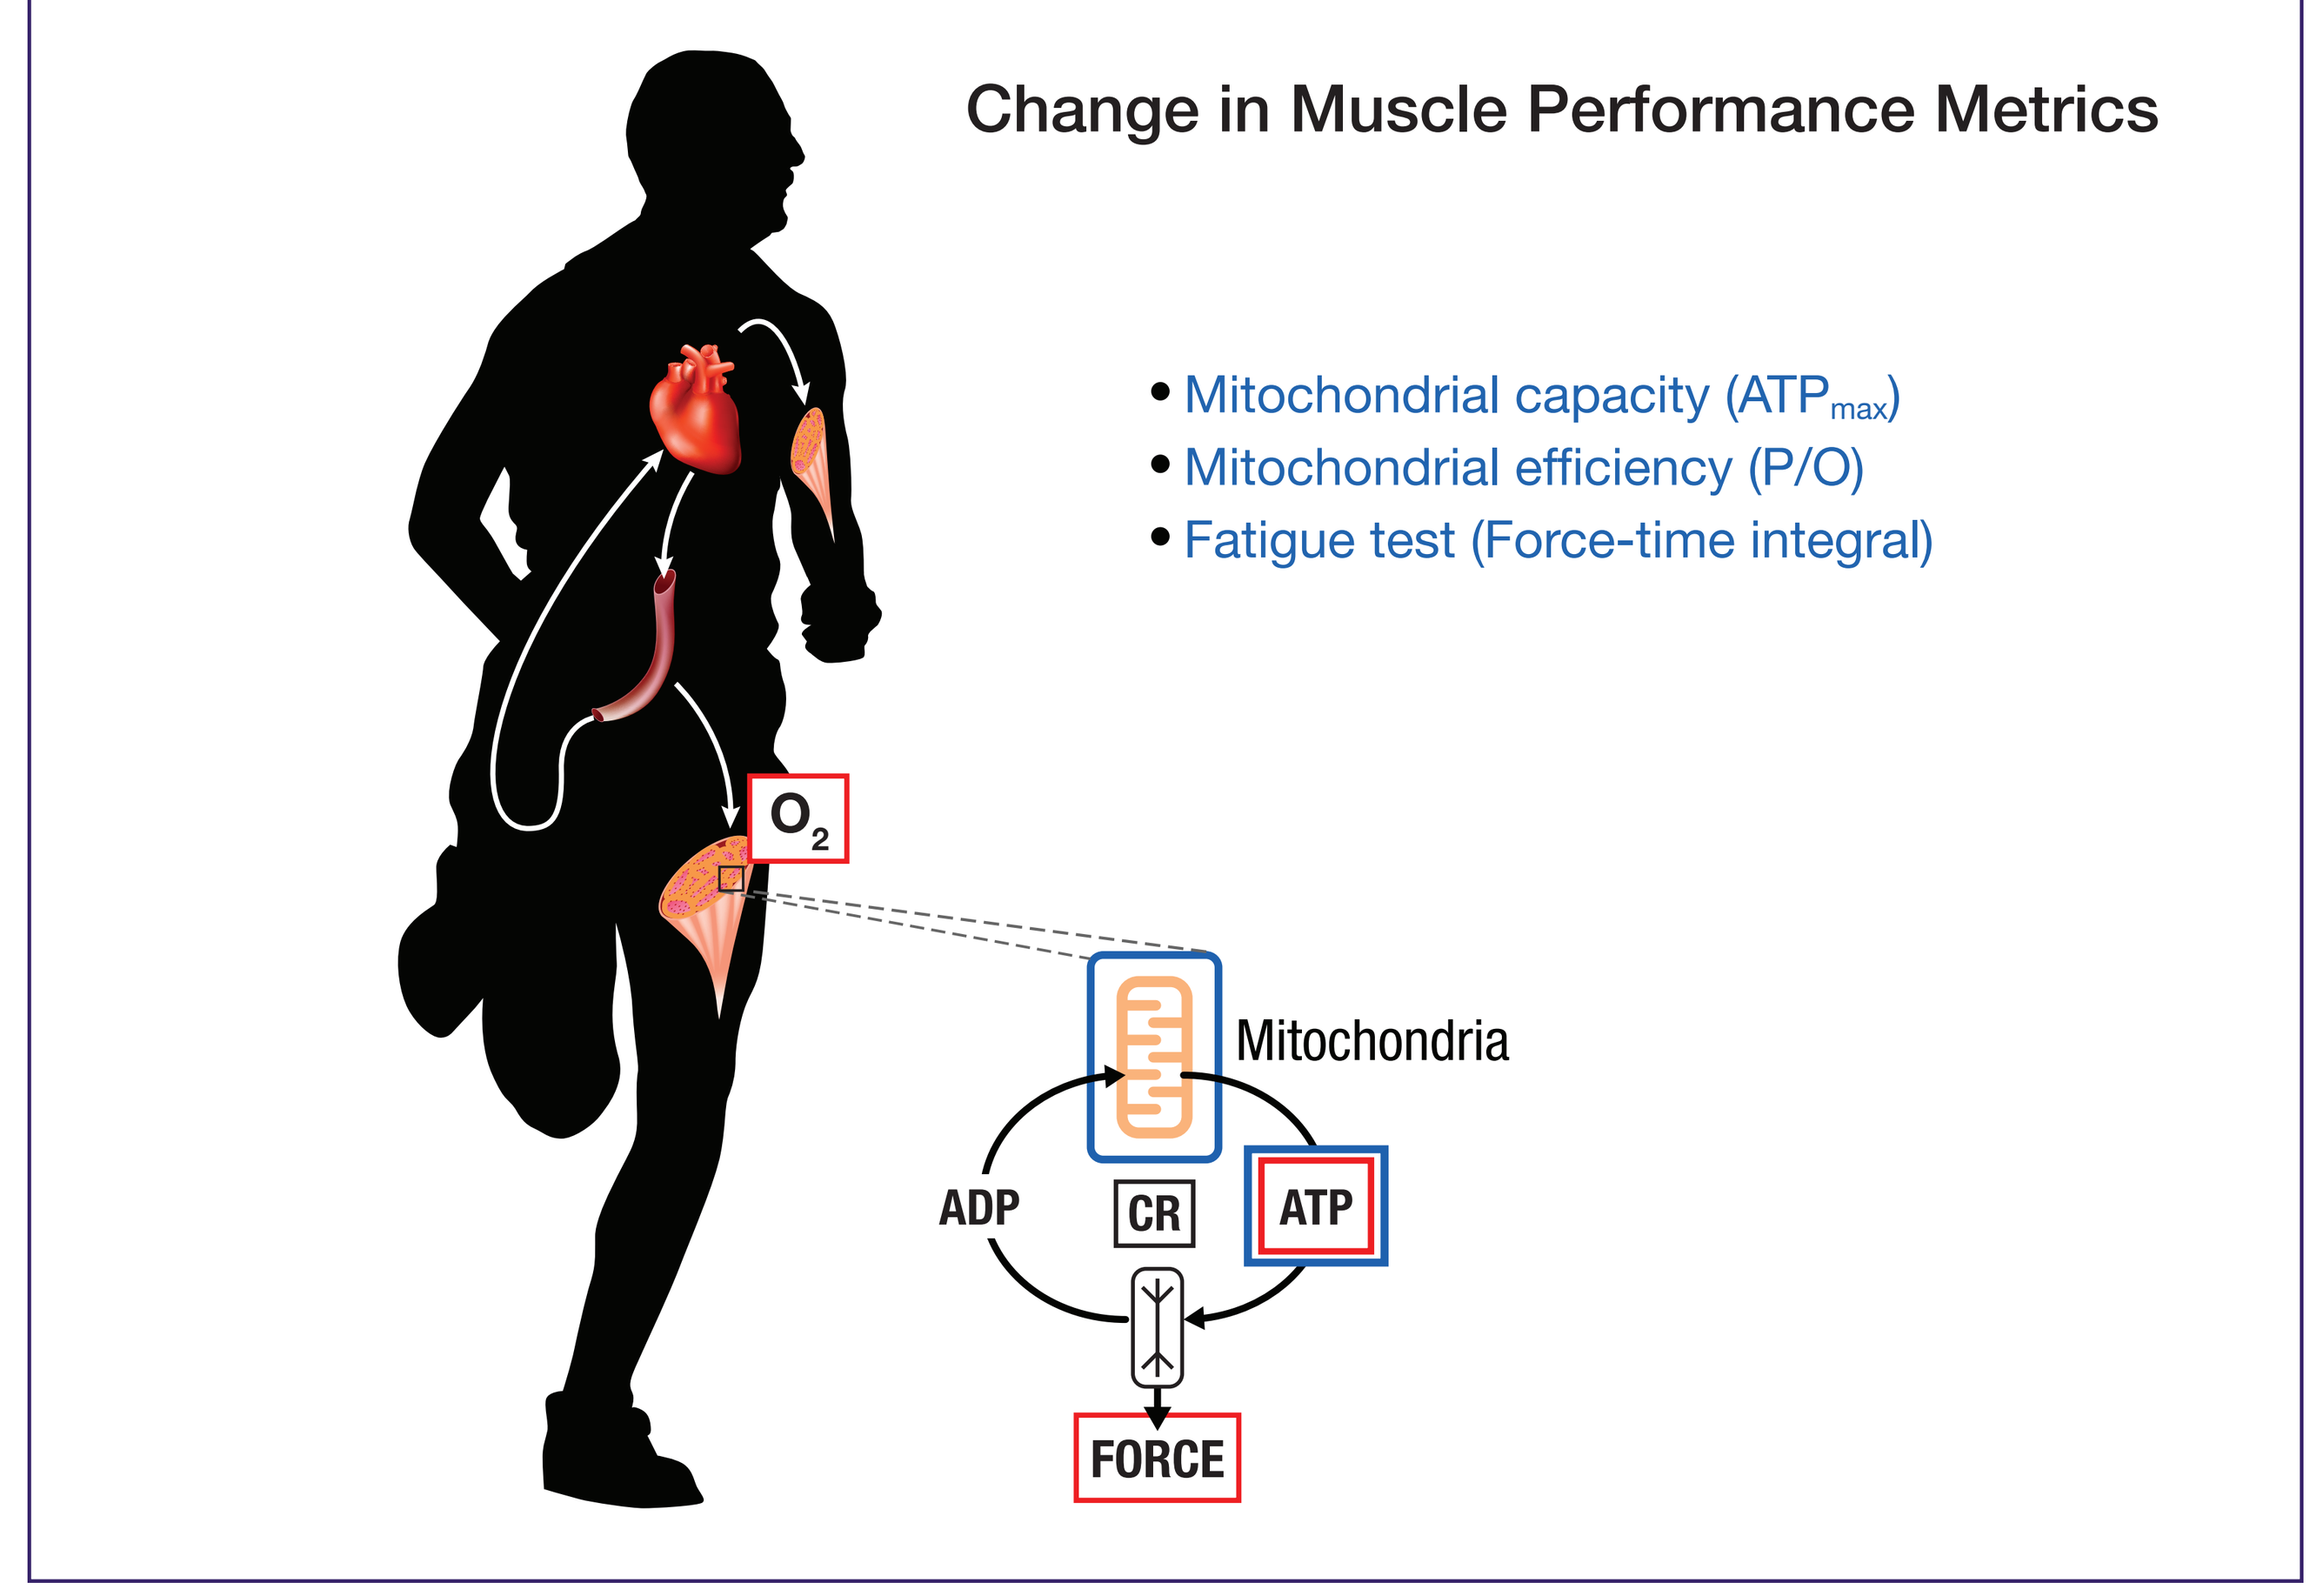
Change in Muscle Performance Metrics
• Mitochondrial capacity (ATP_{max})
• Mitochondrial efficiency (P/O)
• Fatigue test (Force-time integral)
O_2
Mitochondria
ADP
CR
ATP
FORCE

METHODS

Figure 5. Trial Design.

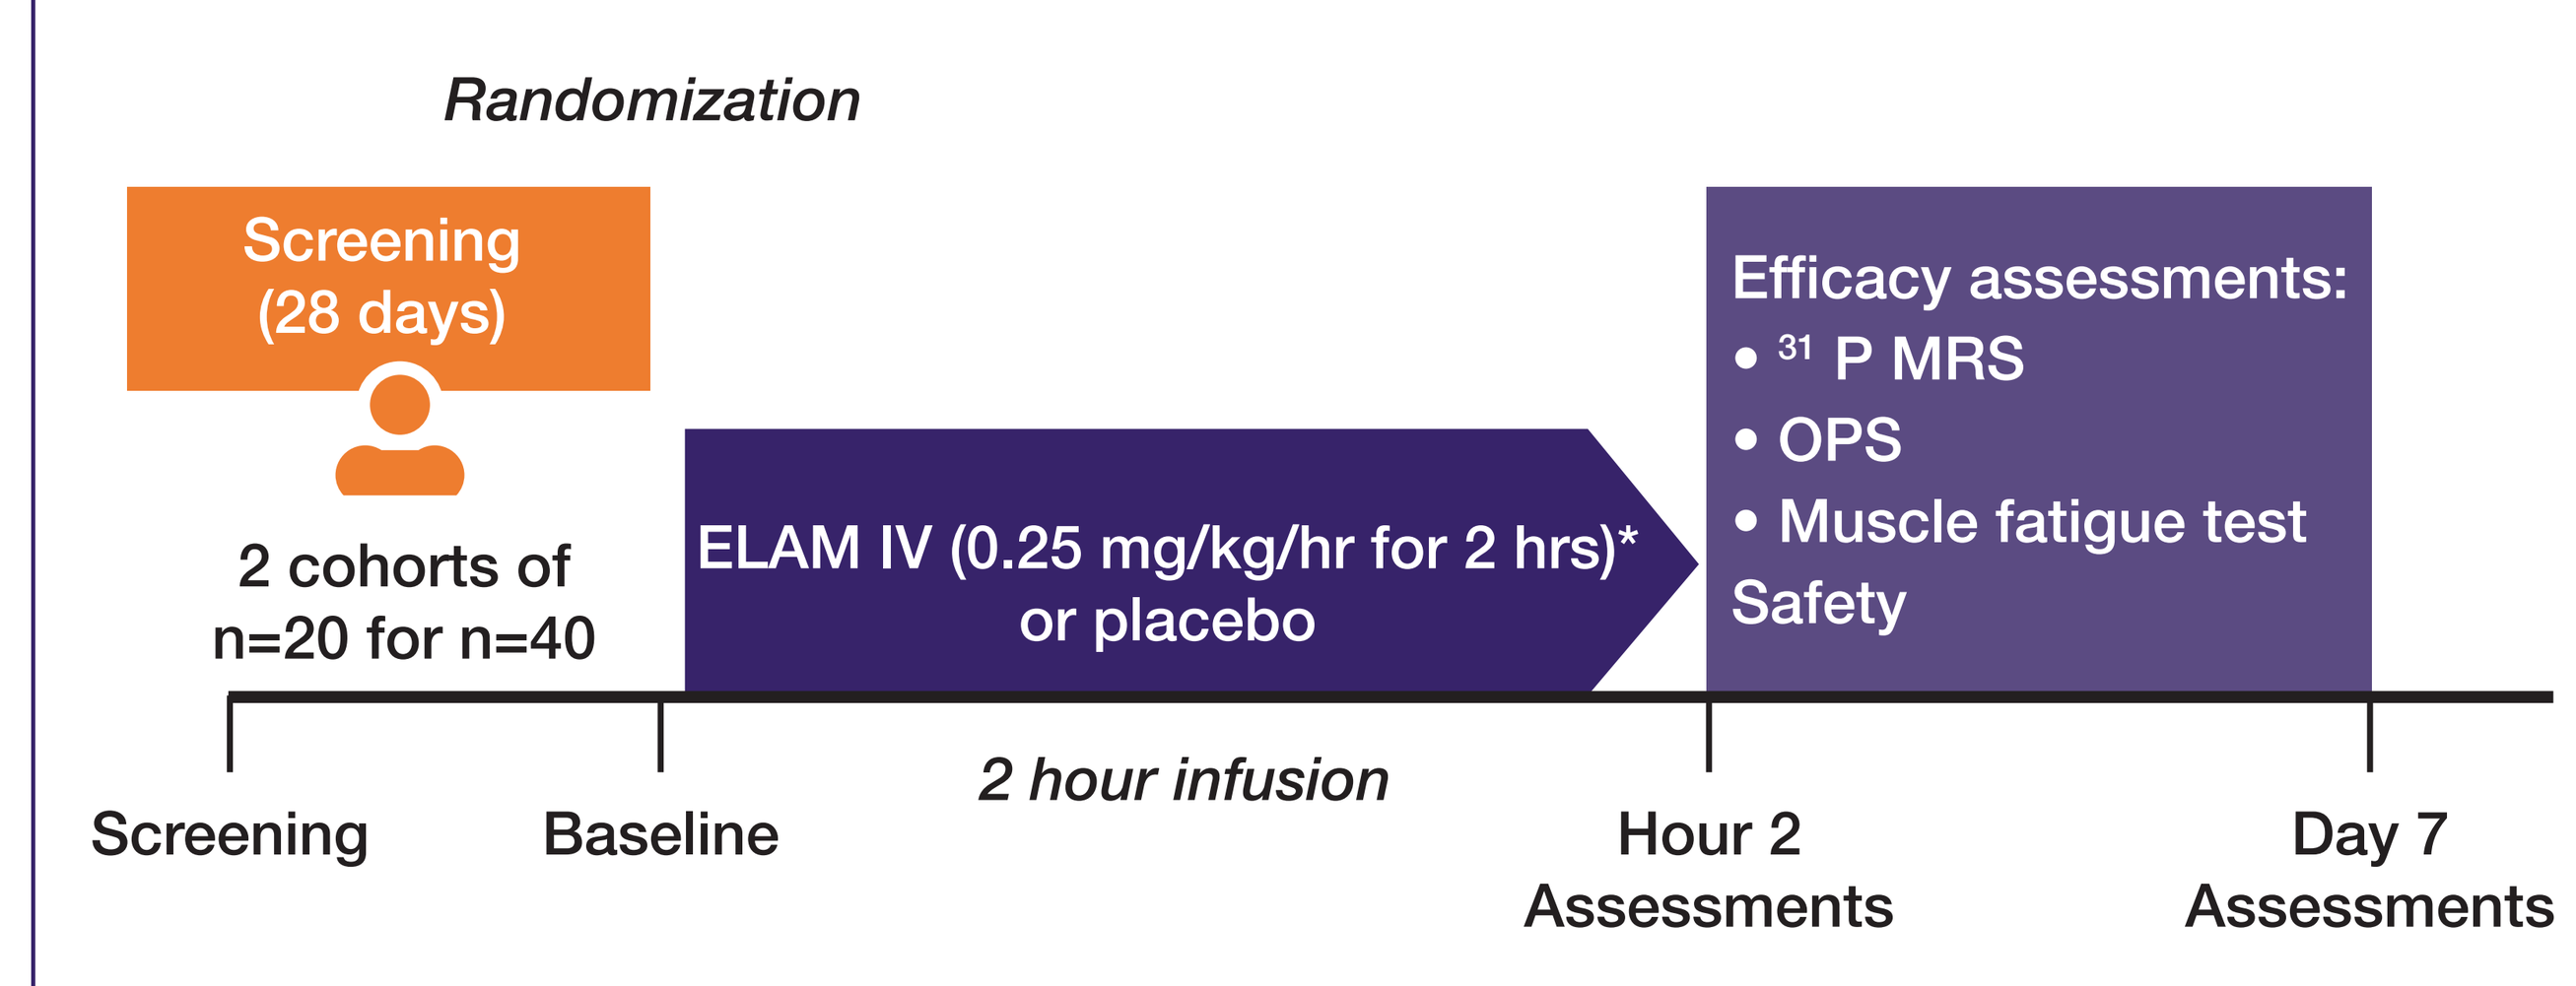
Randomization
Screening (28 days)
2 cohorts of n=20 for n=40
ELAM IV (0.25 mg/kg/hr for 2 hrs)* or placebo
Efficacy assessments:
• [31] P MRS
• OPS
• Muscle fatigue test
Safety
Screening
Baseline
2 hour infusion
Hour 2 Assessments
Day 7 Assessments

*Avg Dose 37 mg

Primary endpoints

- Change from baseline after study drug infusion in the maximum ATP synthetic rate (ATP_{max})
- Actual values and change from baseline were summarized by treatment group
 - The primary analysis set is the intent-to-treat (ITT) population

Secondary endpoints

- Comparisons in change from Baseline between treatment groups for the following hand skeletal muscle energetics and functional properties analyzed in the same manner as the primary efficacy measures were included
 - P/O
 - Muscle force-time-integral
- Change from baseline for the above measures assessed at Hour 2 (2 hours after the start of infusion or end of infusion) and Day 7, except for muscle performance that was also assessed at Day 3
 - Mean change between treatment groups compared in an analysis of covariance (ANCOVA) framework, with Baseline as a covariate

Selected Inclusion Criteria

- Elderly subjects (≥60 yrs of age and ≤85 yrs of age), with a body mass index (BMI) between 16 and 35kg/m² and a baseline 30% reduction in mitochondrial function (n = 38)

Selected Exclusion Criteria

- Subjects were excluded from the study if they were hospitalized within three months prior to screening for a major medical condition, had uncontrolled hypertension, or symptoms of peripheral neuropathy

Safety Assessments

- Treatment-emergent adverse events (TEAEs) were summarized as SAEs and Aes throughout the duration of the study

RESULTS

Figure 6. Change from baseline in ATP_{max} with ELAM versus placebo 2 hrs post-infusion and after 7 days.

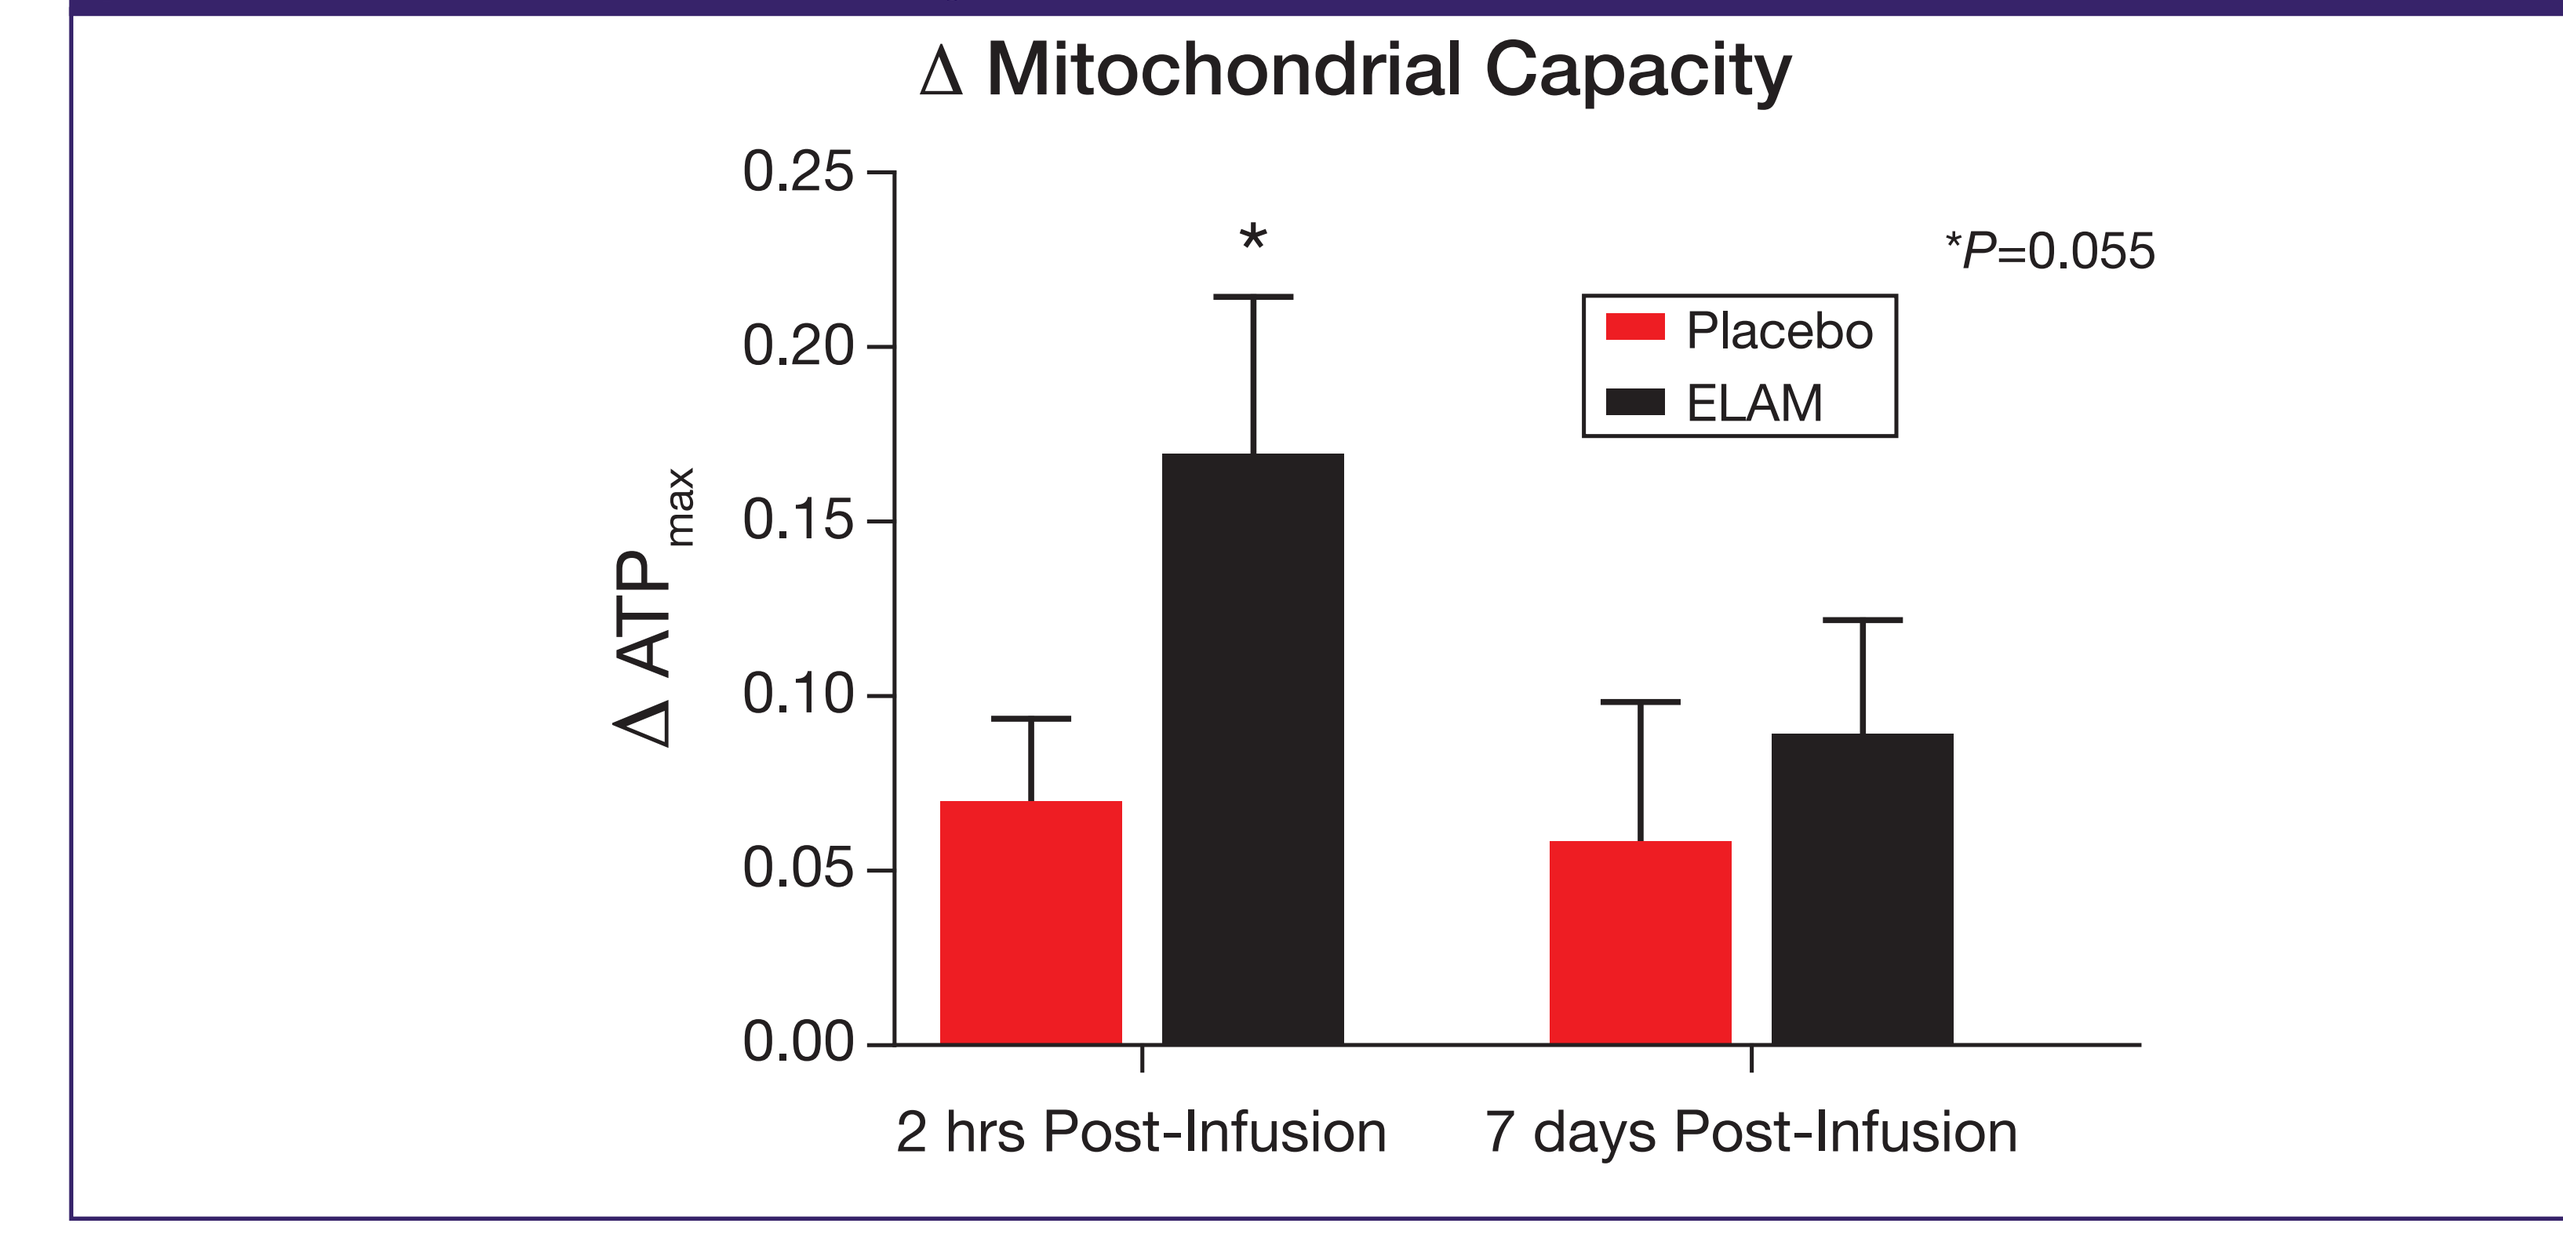
Δ Mitochondrial Capacity
Δ ATP_{max}
0.25
0.20
0.15
0.10
0.05
0.00
*
*P=0.055
Placebo
ELAM
2 hrs Post-Infusion
7 days Post-Infusion

Figure 7. Observed increases in ATP_{max} with ELAM. Comparison to findings from a previous study of elderly patients after 6 months of endurance training.[1]

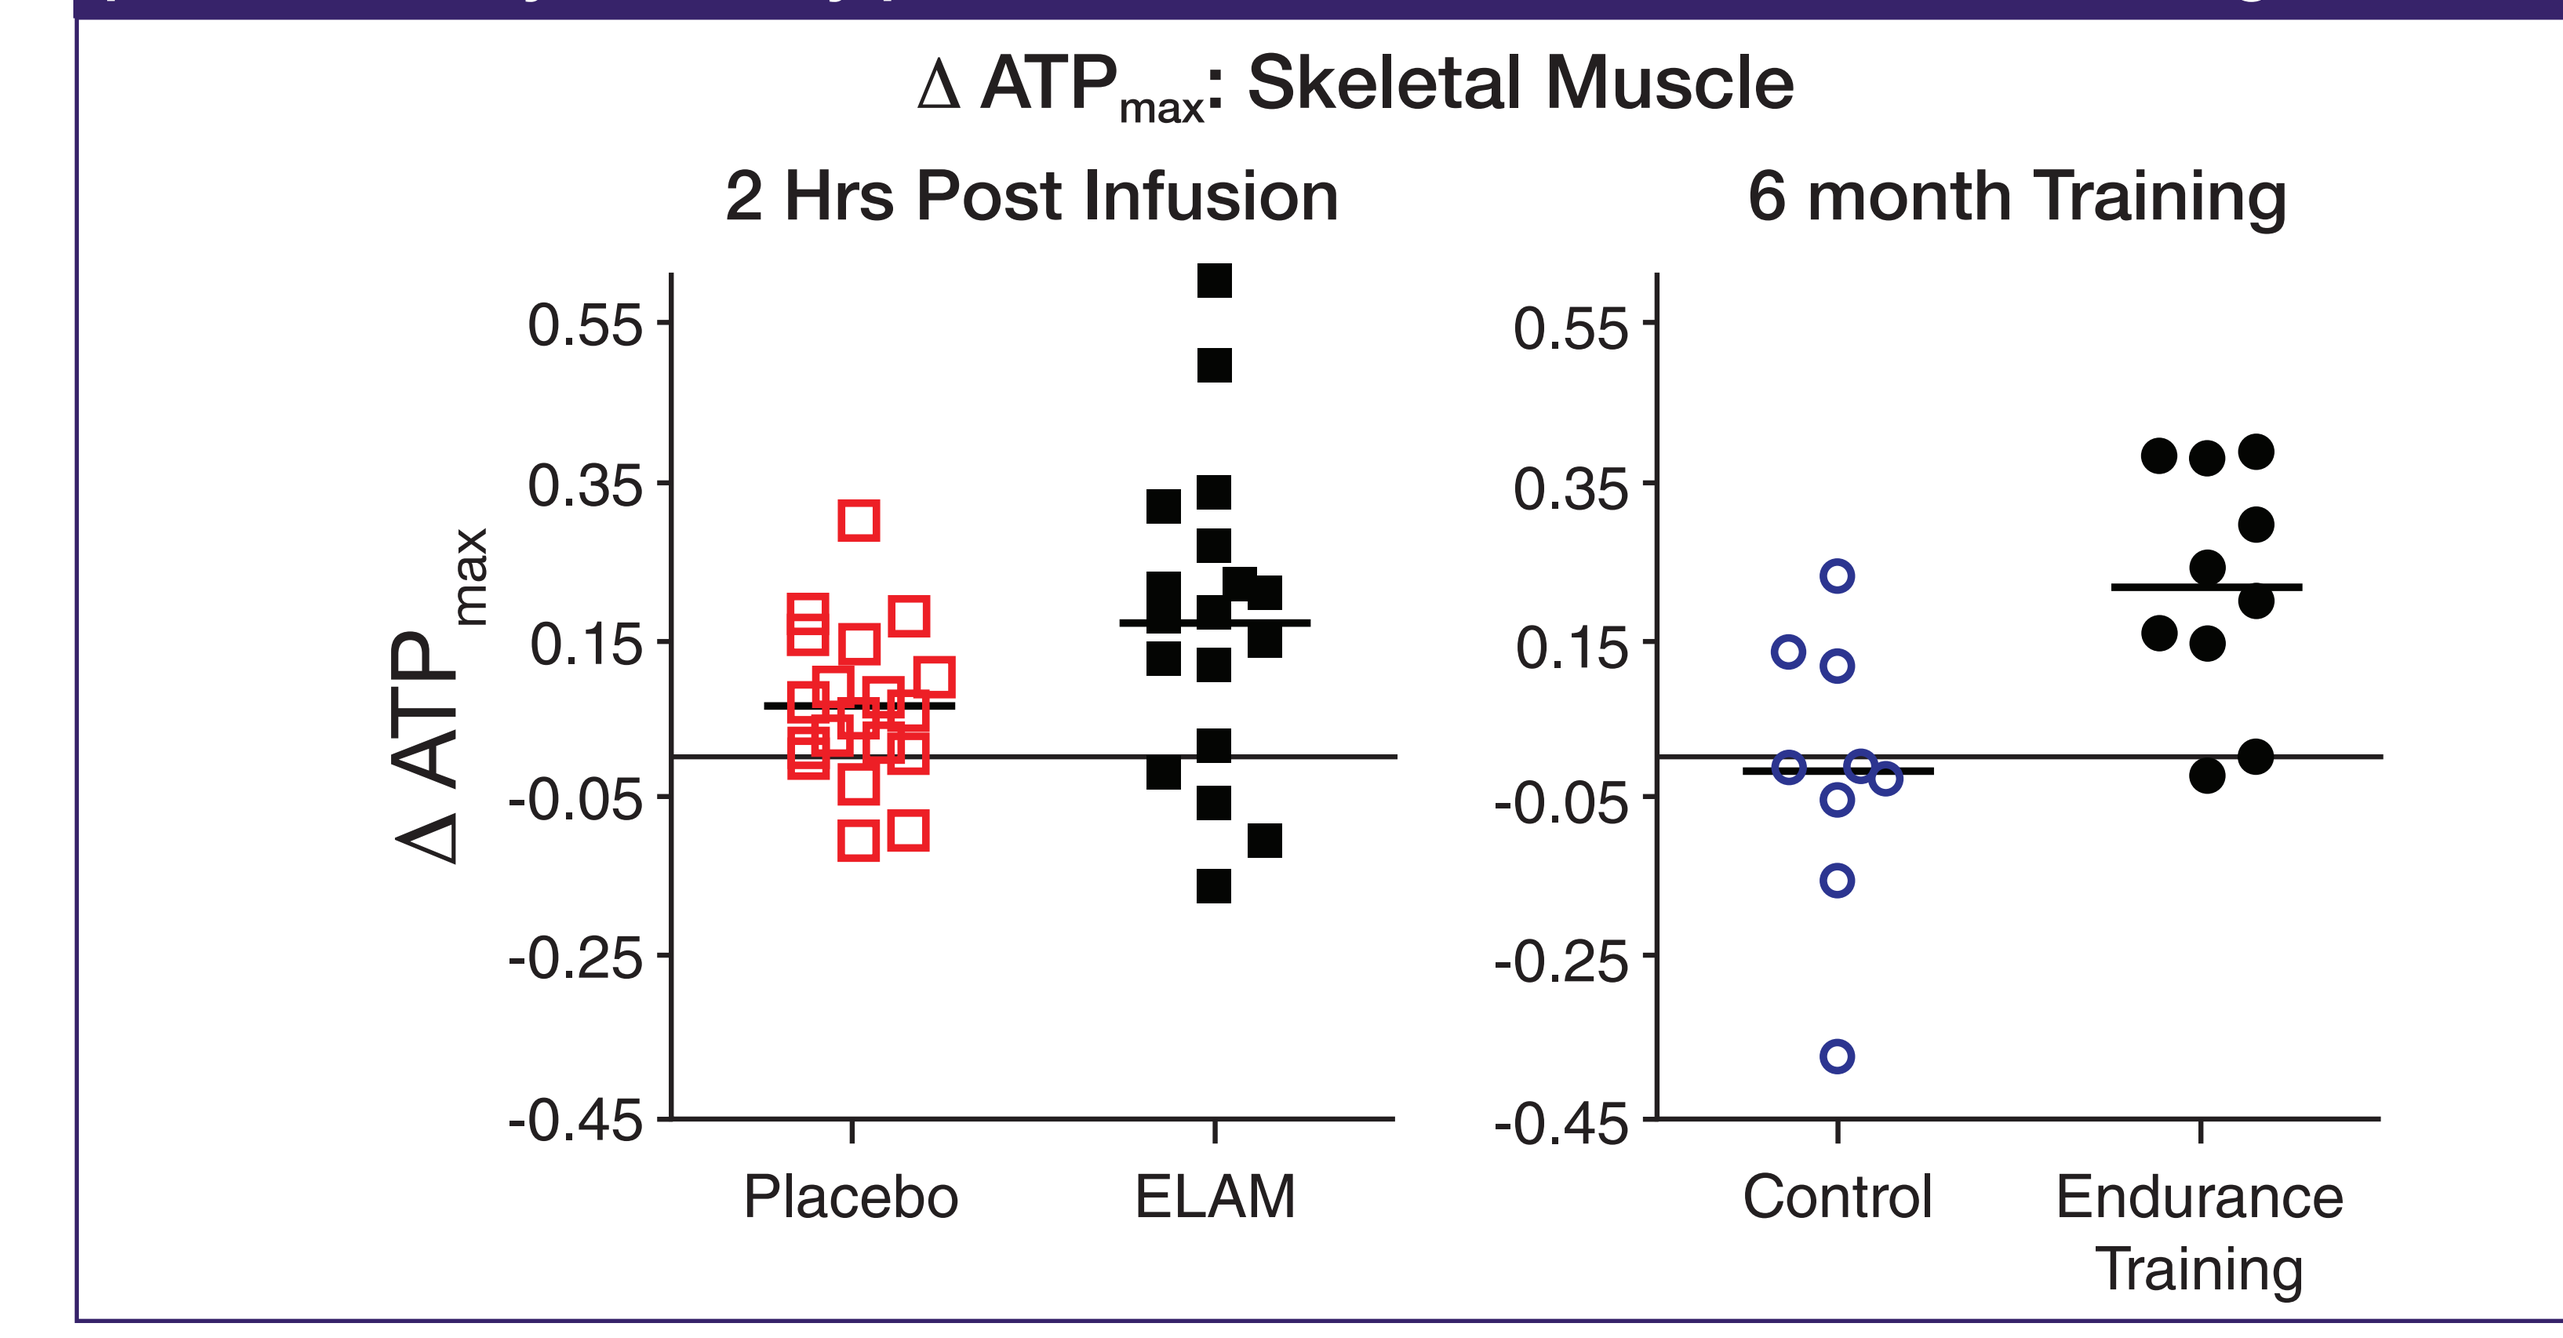
Δ ATP_{max}: Skeletal Muscle
2 Hrs Post Infusion
6 month Training
Δ ATP_{max}
0.55
0.35
0.15
-0.05
-0.25
-0.45
Placebo
ELAM
Control
Endurance Training

- One placebo subject, whose values were determined to be erroneous (albeit after the blind was broken), was dropped from the analysis
- In the study as a whole, higher ATP_{max} values were associated with greater function as measured by Force Time Integral (r=0.267, *p*=0.0041)
- ELAM was well-tolerated; no safety issues were identified

CONCLUSIONS

- ELAM improves mitochondrial function by increasing ATP production after a single infusion
- Reverses mitochondrial dysfunction, O_2 demand and improves sustained exercise performance in the muscle of elderly subjects
- ELAM improvement in ATP_{max} was comparable to ATP_{max} change seen post 6 months of exercise training in this patient population
- ELAM targets the mitochondrial dysfunction that exists in patients with HF, thereby potentially providing improvements in exercise tolerance
- ELAM is an investigational product under study in HF patients

REFERENCES

1. Conley KE, Jubrias SA, Esselman PC. Oxidative capacity and ageing in human muscle. *J Physiol*. 2000 Jul1;526 Pt 1:203–210.
2. Schaefer AM, Phoenix C, Elson JL, McFarland R, Chinnery PF, Turnbull DM. Mitochondrial disease in adults: a scale to monitor progression and treatment. *Neurology*. 2006 Jun 27;66(12):1932–1934.
3. Taivassalo T, Jensen TD, Kennaway N, DiMauro S, Vissing J, Haller RG. The spectrum of exercise tolerance in mitochondrial myopathies: a study of 40 patients. *Brain*. 2003 Feb;126(Pt 2):413–423.
4. Taivassalo T, Haller RG. Implications of exercise training in mtDNA defects--use it or lose it? *Biochim Biophys Acta*. 2004 Dec 6;1659 (2-3):221–231.
5. Pfeffer G, Chinnery P. Diagnosis and treatment of mitochondria myopathies. *Ann Med*. 2013; 45(1):4–16.
6. Gorman GS, Elson JL, Newman J, Payne B, McFarland R, Newton JL, Turnbull DM. Perceived fatigue is highly prevalent and debilitating in patients with mitochondrial disease. *Neuromuscular Disorders*. 2015. 25(7):563–566.
7. Apabhai S, Gorman GS, Sutton L, Elson JL, Plötz T, et al. *Habitual Physical Activity in Mitochondrial Disease*. 2011; 6(7): e22294.
8. Coen PM, Jubrias SA, Distefano G, Amati F, Mackey DC, et al. Skeletal muscle mitochondrial energetics are associated with maximal aerobic capacity and walking speed in older adults. *J Gerontol A Biol Sci Med Sci*. 2013 Apr;68(4):447–455.
9. Birk AV, Chao WM, Bracken C, Warren JD, Szeto HH. Targeting mitochondrial cardiolipin and the cytochrome c/cardiolipin complex to promote electron transport and optimize mitochondrial ATP synthesis. *Br J Pharmacol*. 2014;171(8):2017–2028.
10. Brown DA, Sabbah HN, Shaikh SR. Adapted with permission from: Mitochondrial inner membrane lipids and proteins as targets for decreasing cardiac ischemia/reperfusion injury. *Pharmacol Ther*. 2013 Dec;140(3):258–266.
11. Brown DA, Hale SL, Baines CP, et al. Reduction of early reperfusion injury with the mitochondria-targeting peptide Bendavia. *J Cardiovasc Pharmacol Ther*. 2014;19:121–132.
12. Siegel MP, Kruse SE, Percival JM, et al. Mitochondrial-targeted peptide rapidly improves mitochondrial energetic and skeletal muscle performance in aged mice. *Aging Cell*. 2013;12:763–771.

Acknowledgements:

This study was sponsored by Stealth BioTherapeutics. Medical writing assistance was provided by James A. Shiffer, RPh, Write On Time Medical Communications, LLC