Elamipretide, a mitochondrial-targeted drug, for the treatment of vision loss in dry AMD with high risk drusen: Results of the Phase 1 ReCLAIM Study.

361
A0238

Michael J. Allingham[1], Priyatham S. Mettu[1], Scott W. Cousins[1]
[1]Ophthalmology, Duke University School of Medicine, Durham, NC

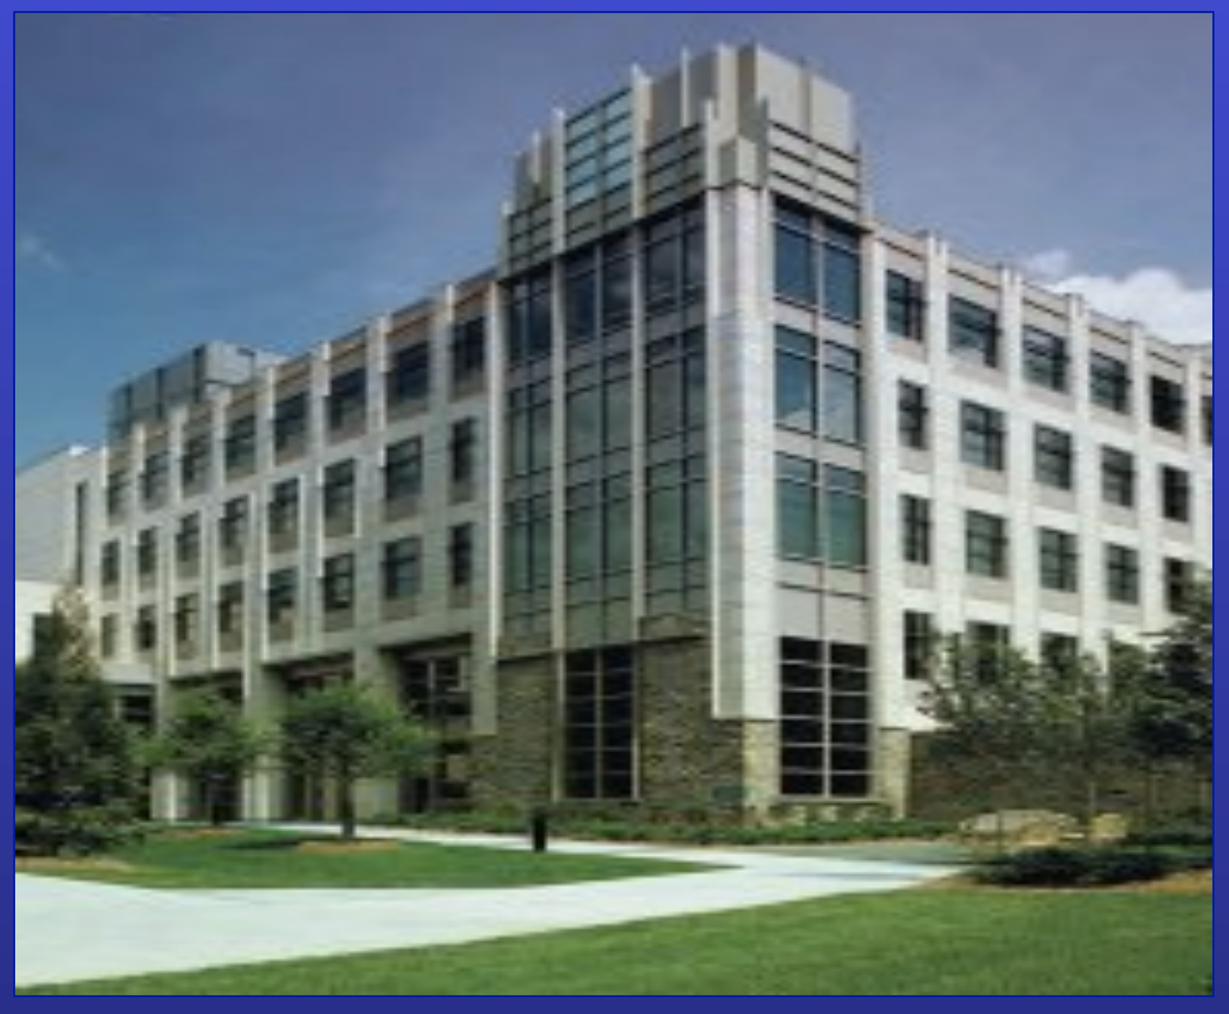

Department of Ophthalmology
Duke University School of Medicine

Abstract

Purpose: To report the results from the high-risk drusen (HRD) subgroup of the ReCLAIM Study, an open-label, phase 1 clinical trial to evaluate safety, tolerability, and efficacy of elamipretide in participants with intermediate age-related macular degeneration (AMD).

Methods: Open-label, phase 1 clinical trial of daily subcutaneous elamipretide (40 mg) for 24 weeks in individuals with dry AMD. For the pre-defined HRD subgroup, HRD was defined as presence of either at least 1 large (≥ 125 μm) druse or multiple medium-size (between 63 and 124 μm) drusen. Eyes with any geographic atrophy were excluded from the HRD subgroup. A single eye of each participant was eligible if ETDRS best-corrected visual acuity (BCVA) was ≥ 55 letters with low luminance VA (LLVA) deficit > 5 letters. LLVA was measured as best-corrected ETDRS VA through a log 2 neutral density filter and LLVA deficit was defined as the difference between BCVA and LLVA. Safety and visual function assessments occurred every four weeks, with outcomes assessed at week 24.

Results: Subcutaneous elamipretide was safe and well tolerated with no treatment-related serious adverse events. Among participants in the HRD subgroup (n=21, age 70.9, 61.9% female), mean BCVA at baseline was 79.6 ± 7.4 letters, with mean increase of 3.6 ± 6.4 letters (p=0.025) at week 24. Mean baseline LLVA was 63.7 ± 10.0 letters, with mean increase of 5.6 ± 7.8 letters (p=0.006). Mean best-corrected reading acuity under standard lighting conditions (BCRA) (tested using MN read acuity chart) at baseline was logMAR 0.01 ± 0.18, with mean increase of -0.11 ± 0.15 (p=0.005), equivalent to an approximately 1-line gain. Mean low luminance reading acuity through log 2 neutral density filter (LLRA) was logMAR 0.39 ± 0.23 at baseline, with mean increase of -0.28 ± 0.17 (p=0.0001), equivalent to an approximately 3-line gain.

Conclusion: Elamipretide, a mitochondrial-targeted drug, is safe and well-tolerated and may improve vision in patients with intermediate (dry) AMD manifest as HRD.

Introduction

Figure 1: Preclinical rationale

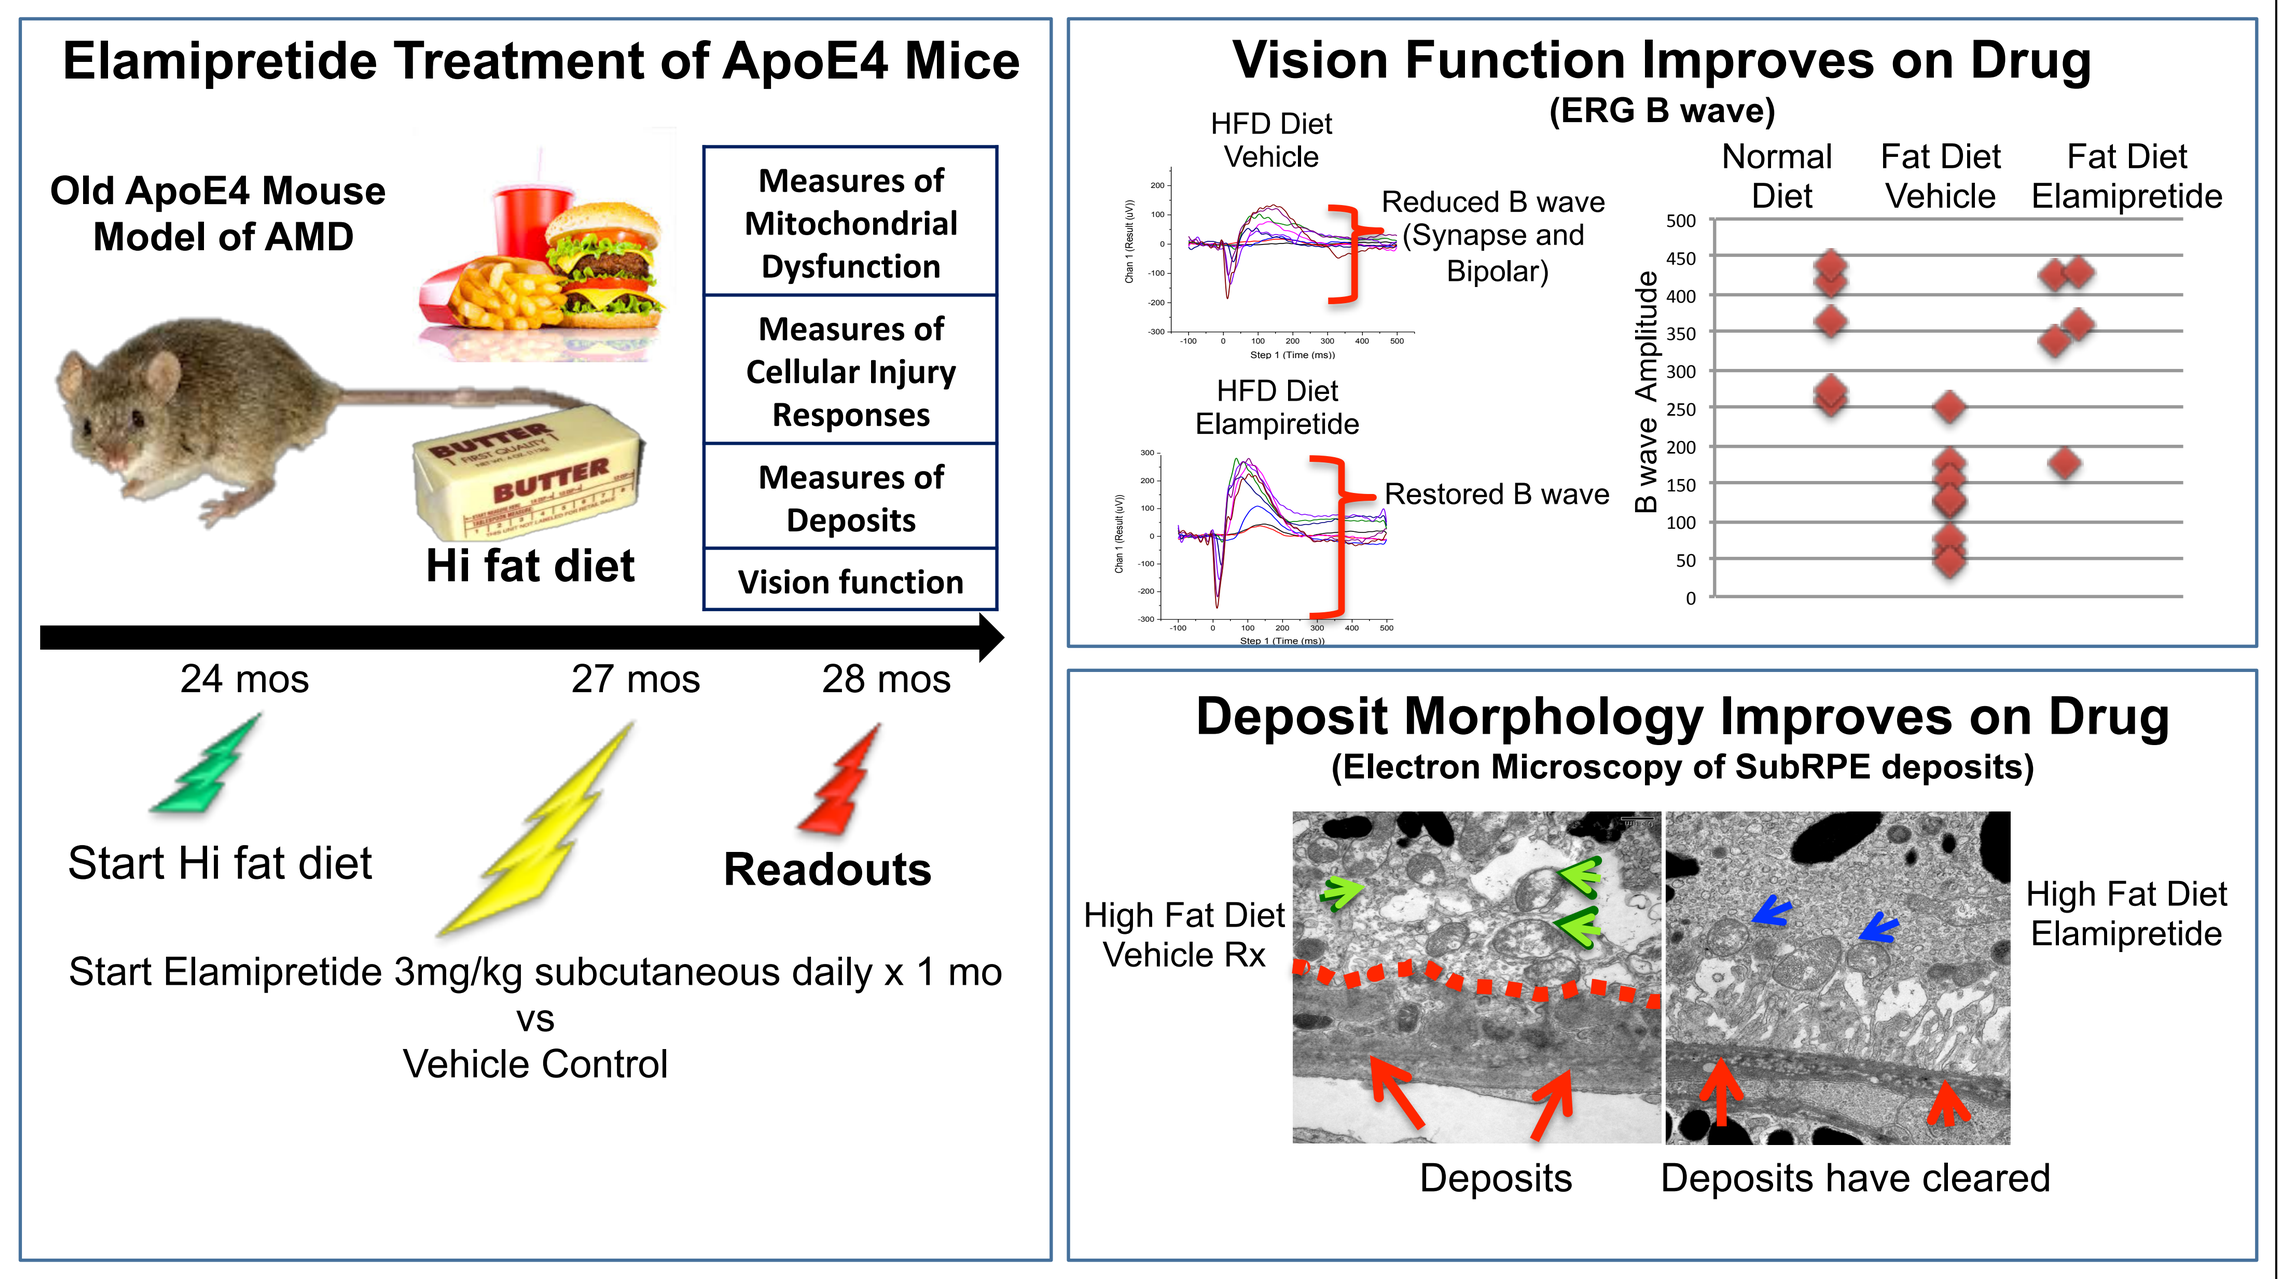
Elamipretide Treatment of ApoE4 Mice
Old ApoE4 Mouse Model of AMD
Hi fat diet
Measures of Mitochondrial Dysfunction
Measures of Cellular Injury Responses
Measures of Deposits
Vision function
24 mos
27 mos
28 mos
Start Hi fat diet
Readouts
Start Elamipretide 3mg/kg subcutaneous daily x 1 mo
vs
Vehicle Control
Vision Function Improves on Drug
(ERG B wave)
HFD Diet Vehicle
Reduced B wave (Synapse and Bipolar)
HFD Diet Elampiretide
Restored B wave
Normal Diet
Fat Diet Vehicle
Fat Diet Elamipretide
B wave Amplitude
Deposit Morphology Improves on Drug
(Electron Microscopy of SubRPE deposits)
High Fat Diet Vehicle Rx
High Fat Diet Elamipretide
Deposits
Deposits have cleared

Figure 1: One month of elamipretide treatment reversed existing visual dysfunction and subRPE deposits in the ApoE4 mouse model of dry AMD.

Figure 2: Study design

Screen

BL W1 W4 W8 W12 W16 W20 W24

W28 washout

High Risk Drusen + BCVA ≥ 55 Ltrs + > 5 Ltrs Low luminance deficit n=21

High Risk Drusen + Noncentral GA + BCVA ≥ 55 Ltrs + > 5 Ltrs Low luminance deficit n=19

Elamipretide 40 mg subcutaneous once daily

Endpoints
Primary Endpoint: Safety
- Adverse event attributed to drug

Exploratory Endpoints: Efficacy, change in
- Best corrected visual acuity (BCVA)
- Low luminance visual acuity (LLVA)
- Low luminance reading acuity (LLRA)

- Dark adaptation (DA)
- Low Luminance Questionnaire (LLQ)
- NEI Visual Function Questionnaire (VFQ)
- Drusen volume*
- Fundus hyperautofluorescence*
- Cone light sensitivity (microperimetry)*

Figure 2: Subjects meeting inclusion criteria above were treated for 24 weeks with 40mg elamipretide administered subcutaneously each day. The primary endpoint was safety; exploratory endpoints are above. Exclusion criteria included: AMD with central GA, neovascular AMD, macular atrophy due to causes other than AMD, diabetic retinopathy, other macular pathology, prior vitreoretinal surgery, vitreous hemorrhage, uncontrolled glaucoma, advanced guttae, visually significant cataract, significant posterior capsular opacity, aphakia, significant keratopathy, ocular incisional surgery within 3 months of the baseline visit, immunocompromised state or receiving systemic immunosuppression, estimated glomerular filtration rate < 30 mL/minute.

Figure 3: Subject demographics

	NCGA (n=19)	HRD (n=21)	Total (N=40)
Age, years			
Mean (SD)	76.0 (8.22)	70.9 (8.54)	73.3 (8.67)
Median	74.7	69.3	72.8
Minimum, maximum	64, 96	59, 87	59, 96
Sex, n (%)			
Male	8 (42.1)	8 (38.1)	16 (40.0)
Female	11 (57.9)	13 (61.9)	24 (60.0)
Ethnicity, n (%)			
Hispanic or Latino	1 (5.3)	1 (4.8)	2 (5.0)
Not Hispanic or Latino	18 (94.7)	20 (95.2)	38 (95.0)
Race, n (%)			
White	19 (100.0)	21 (100.0)	40 (100.0)
Smoking status, n (%)			
Never smoker	8 (42.1)	13 (61.9)	21 (52.5)
Former smoker	11 (57.9)	8 (38.1)	19 (47.5)
Current smoker	0	0	0

Results

Figure 4: Elamipretide safety data

	NCGA (n=19)	HRD (n=21)	Total (N=40)
Enrolled	19	21	40
Completed study, n (%)	15 (78.9)	18 (85.7)	33 (82.5)
Withdrawn early from the study, n (%)	4 (21.1)	3 (14.3)	7 (17.5)
Reasons for early withdrawal, n (%)			
AE (Injection Site Reactions)	2 (10.5)	1 (4.8)	3 (7.5)
Converted to NV AMD	1 (5.3)	0	1 (2.5)
Withdrawal by subject	1 (5.3)	1 (4.8)	2 (5.0)
Other	0	1 (4.8)	1 (2.5)

Figure 4: Elamipretide was well tolerated in general with no serious adverse events in any subject. The most common AE was injection sight reaction (pruritis, erythema, bruising, urticaria). All AEs were mild to moderate. One subject in the HRD group discontinued study drug due to injection sight reaction which failed to resolve with topical treatment.

Figure 5: Change in best corrected VA

Time Point	High Risk Drusen	n
Baseline	**79.4 ± 7.4** (~20/25)	21
Week 24	**82.0 ± 6.9** (~20/20)	19
Change BL-Wk 24	**3.6 ± 6.4**	**p=0.025**

Figure 5: BCVA was measured using standard ETDRS chart in normal lighting conditions. Change in baseline BCVA from baseline to week 24 showed statistically significant improvement, possibly limited by ceiling effect of good starting BCVA in the HRD subgroup.

Figure 6: Change in low luminance VA

Time Point	High Risk Drusen	n
Baseline	**63.7 ± 10.0** (~20/63 ± 20/40-20/100)	21
Week 24	**68.4 ± 11.5** (~20/50)	19
Change BL-Wk 24	**5.6 ± 7.8**	p=0.006

Figure 6: LLVA was measured as for BCVA except subjects viewed chart through a 2 log ND filter. Change in baseline LLVA from baseline to week 24 showed statistically significant improvement.

Figure 7: Change in normal luminance reading acuity

Time Point	High Risk Drusen Disease Group	n
Baseline	**0.01 ± 0.18**	21
Week 24	**-0.08 ± 0.19**	19
Change BL-Wk 24	**-0.11 ± 0.15**	p=0.005

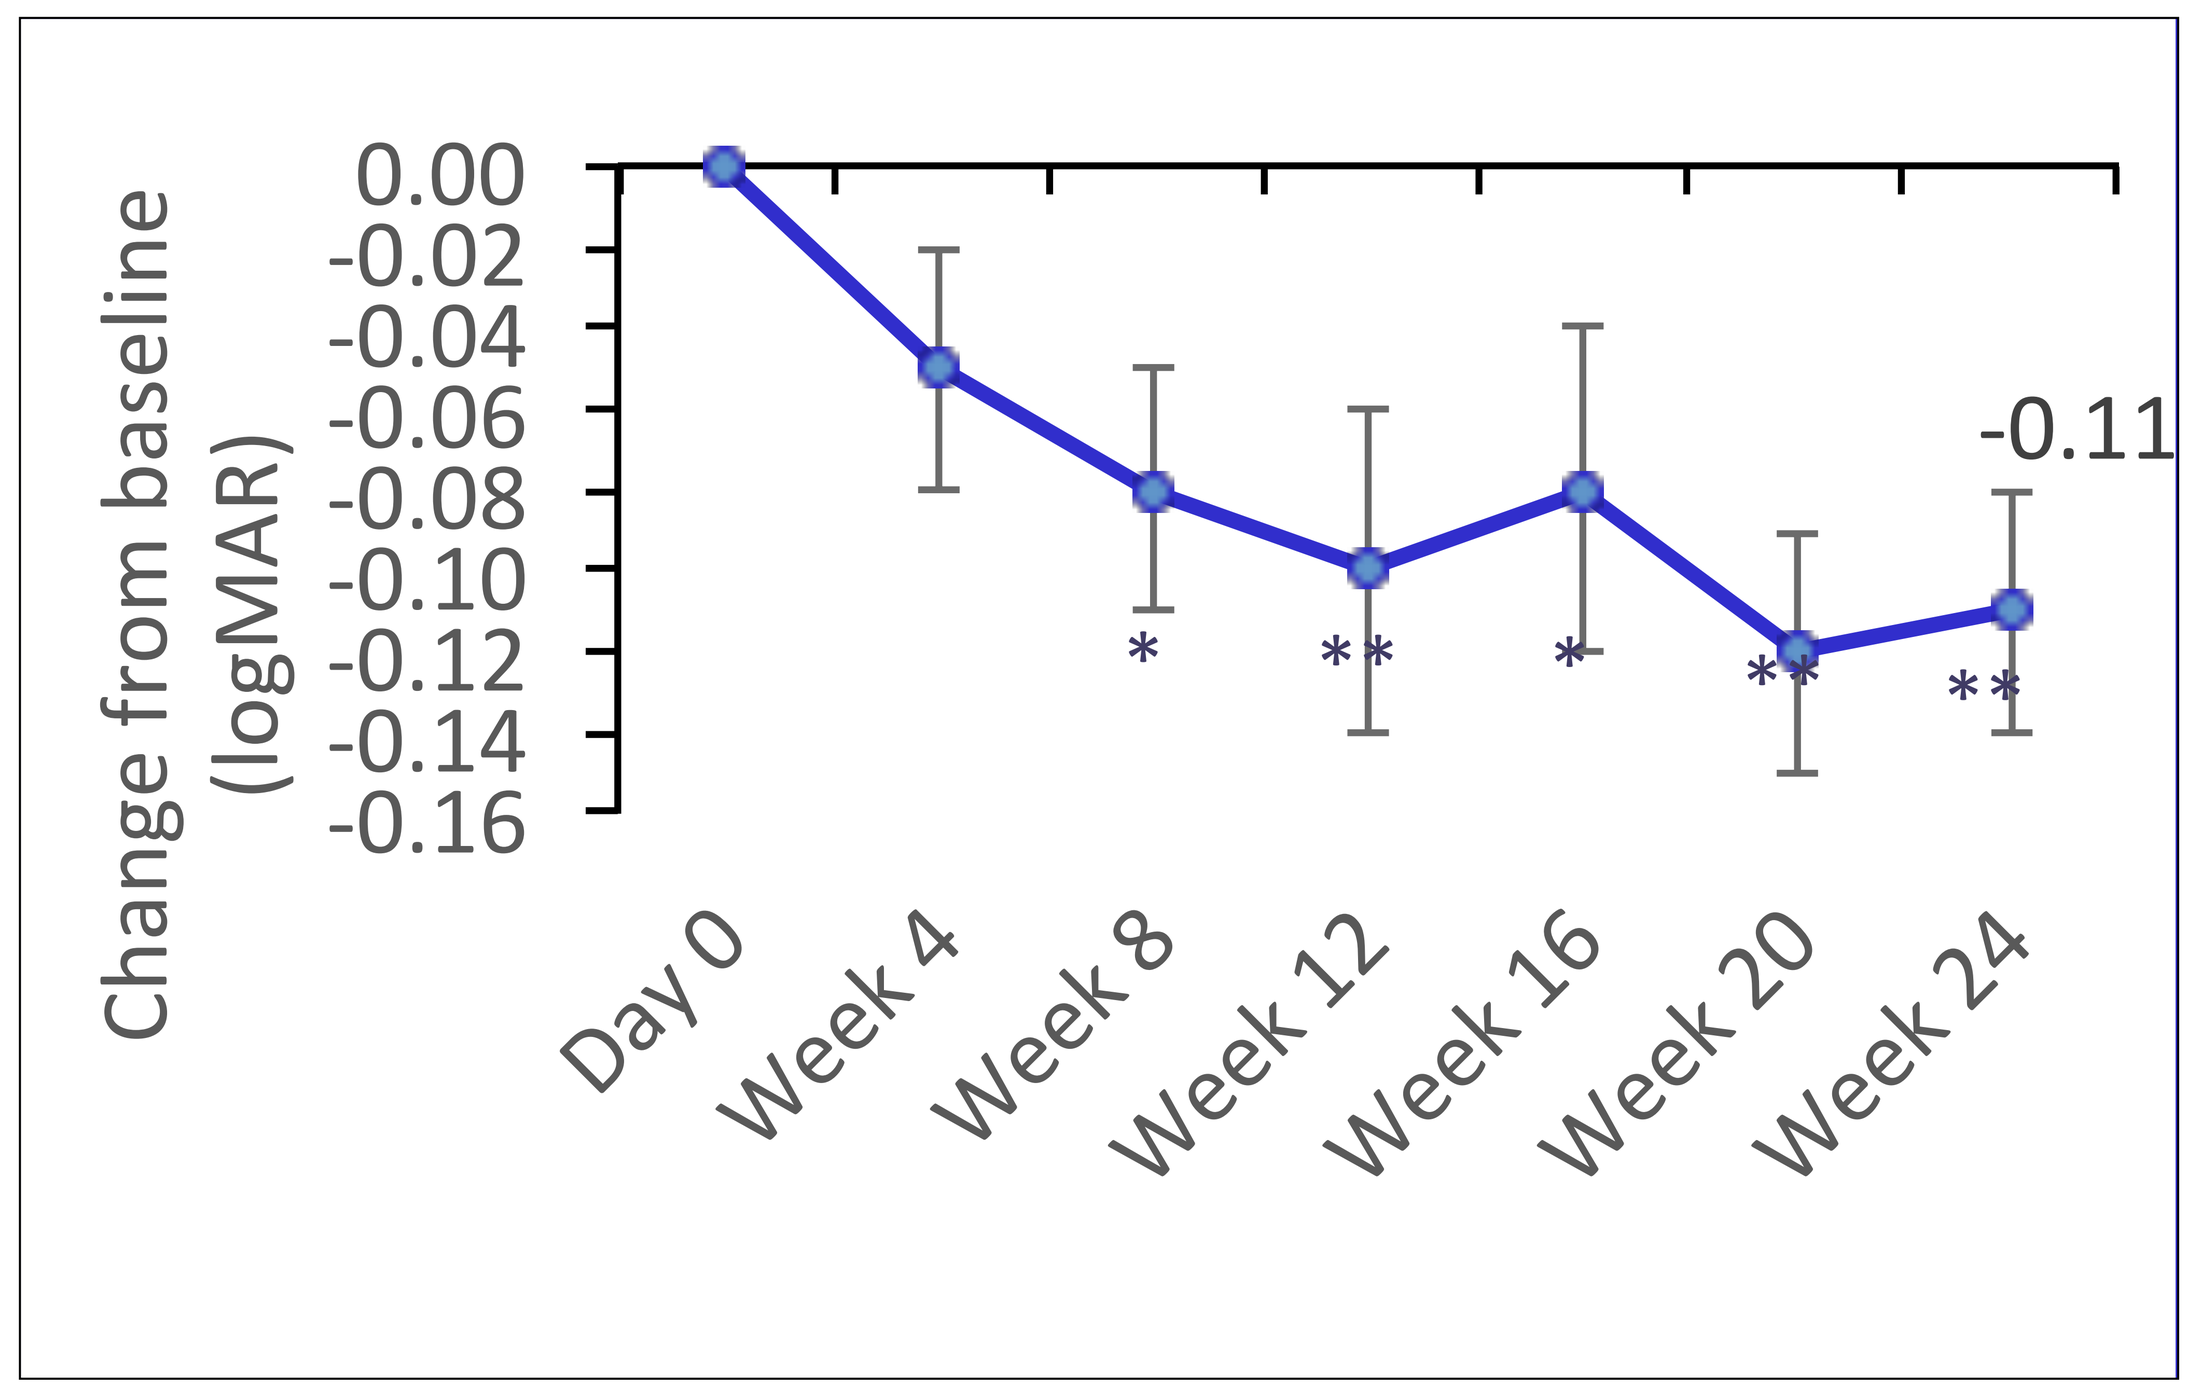
0.00
-0.02
-0.04
-0.06
-0.08
-0.10
-0.12
-0.14
-0.16
Change from baseline (logMAR)
-0.11
*
**
*
**
**
Day 0
Week 4
Week 8
Week 12
Week 16
Week 20
Week 24

Figure 7: Assessment of NLRA was performed in standardized illumination using several different standard MNREAD charts with charts changed each visit to prevent a learning effect. Results were recorded as the smallest font size read correctly with ≤1 word mistake within 30 seconds. Mean improvement in normal luminance reading acuity was statistically significant and equivalent to approximately a 1 line gain. (* p<0.05, ** p<0.01)

Figure 8: Change in low luminance reading acuity

Time Point	High Risk Drusen Disease Group	n
Baseline	**0.39 ± 0.23**	21
Week 24	**0.11 ± 0.21**	19
Change BL-Wk 24	**-0.28 ± 0.17**	p=0.0001

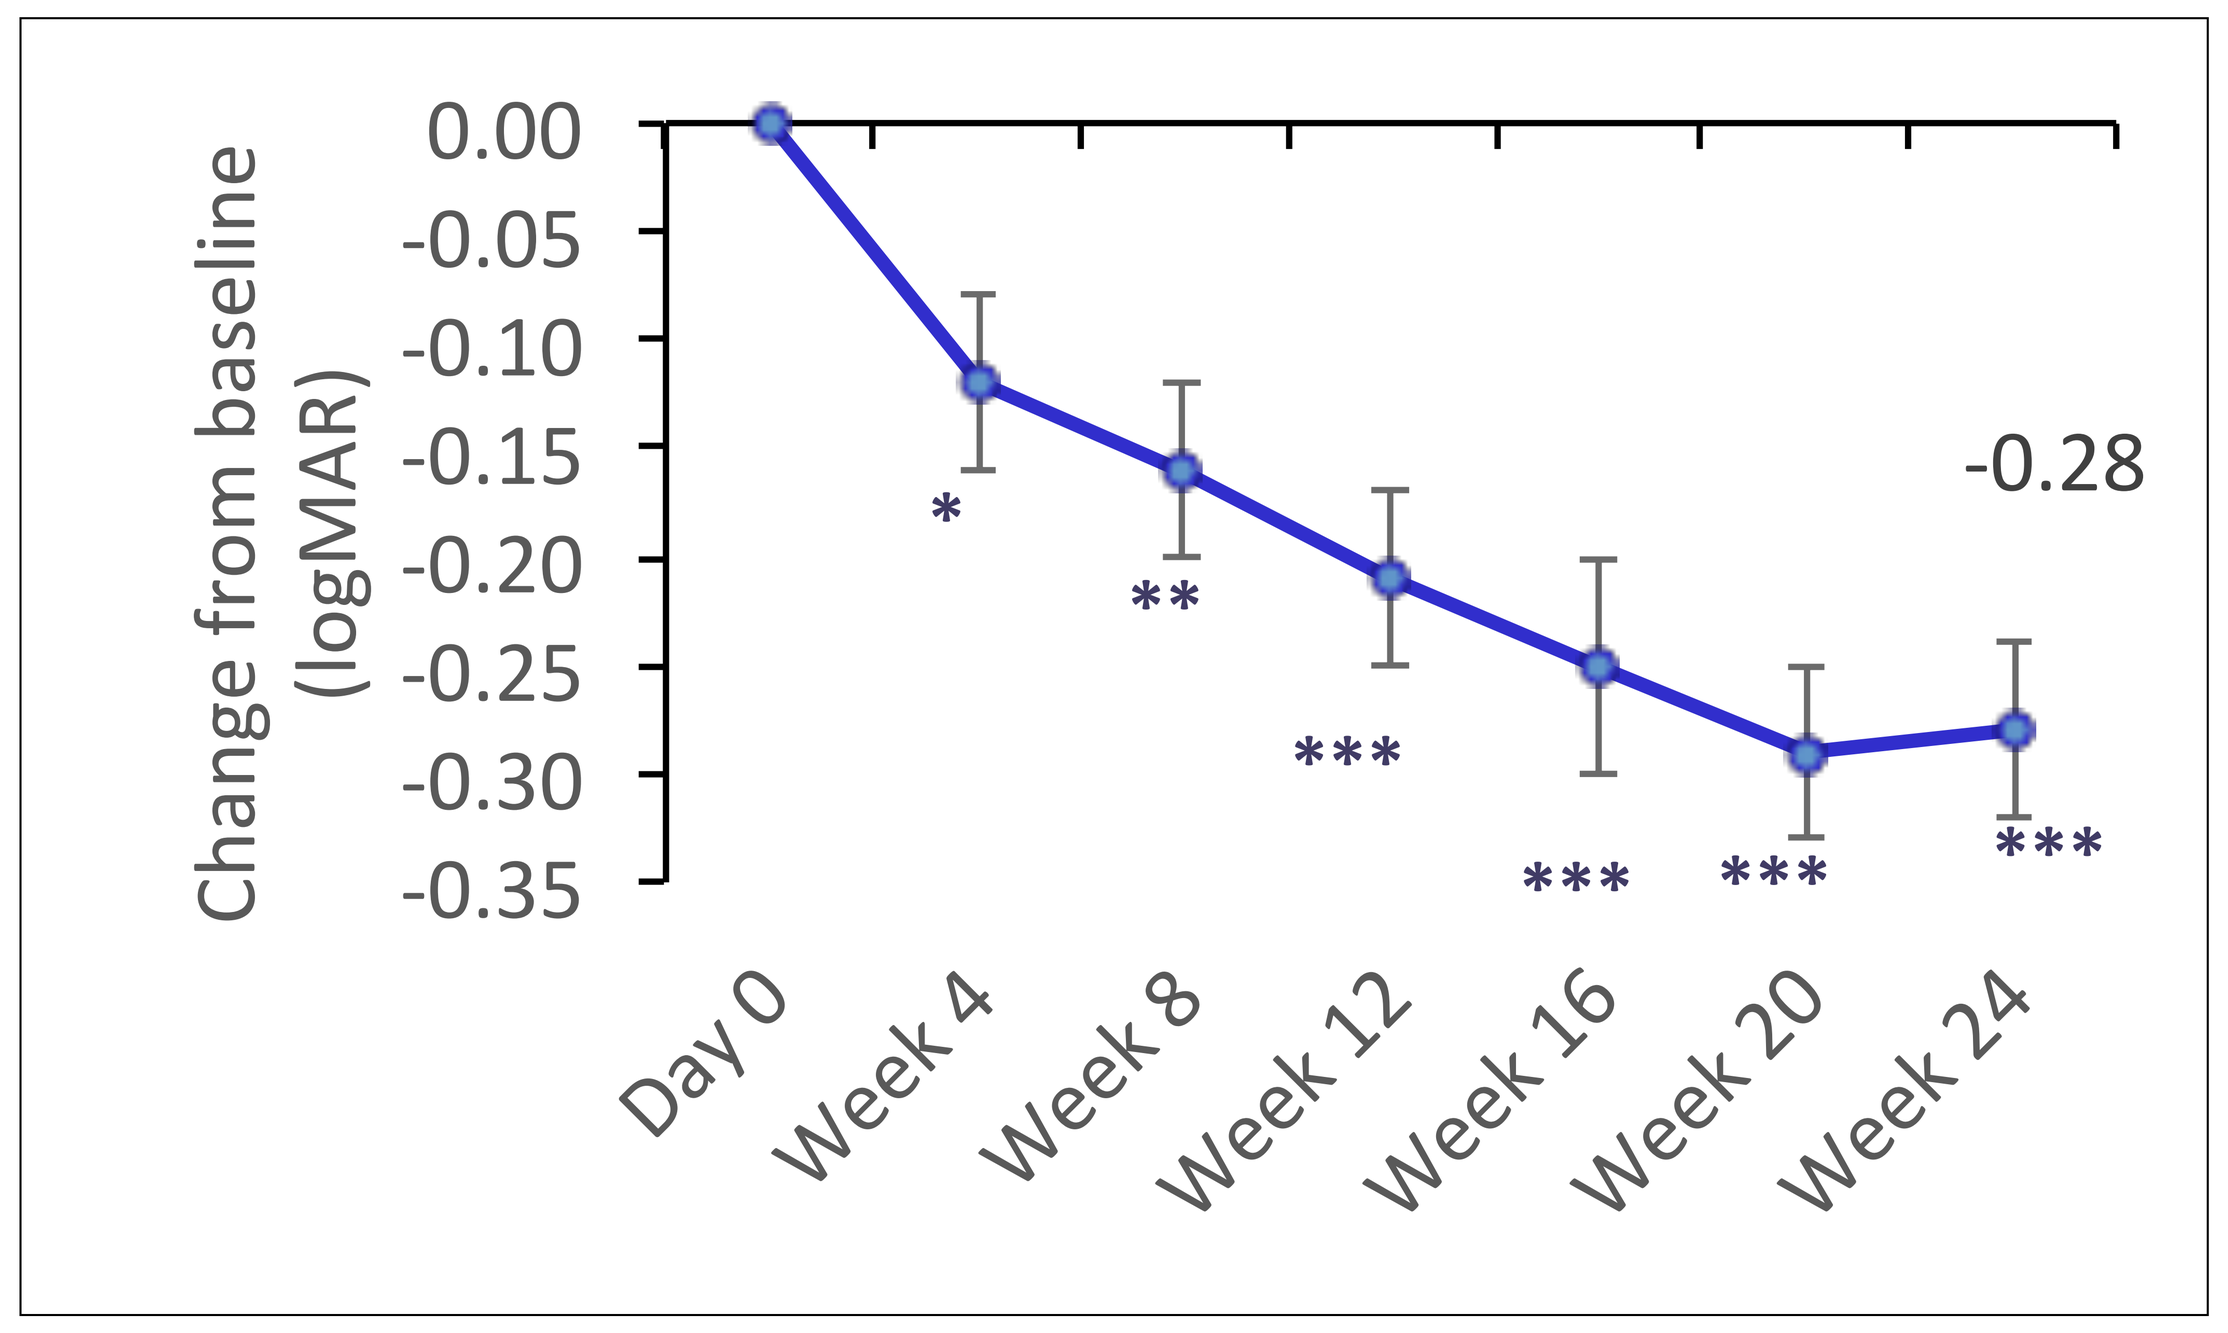
0.00
-0.05
-0.10
-0.15
-0.20
-0.25
-0.30
-0.35
Change from baseline (logMAR)
-0.28
*
**

Day 0
Week 4
Week 8
Week 12
Week 16
Week 20
Week 24

Figure 8: LLRA was performed in a similar fashion as NLRA except that a log 2.0 neutral density filter was added to trial frames with best-corrected near acuity lenses to replicate low-luminance conditions. Mean improvement in LLRA was statistically significant and equivalent to approximately a 3 line gain. (* p<0.05, ** p<0.01, *** p<0.001)

Conclusions

- Elamipretide was safe and well tolerated in subjects with high risk drusen.
- Elamipretide may improve visual function in subjects dry AMD manifest as high risk drusen

Future Directions

- **A phase 2B international, randomized, placebo controlled clinical trial is underway.**

Research Support and Disclosure:
All authors have received financial support and act as consultants for Stealth BioTherapeutics. This clinical trial was funded by a grant to Duke Eye Center from Stealth BioTherapeutics

Author contact: mike.allingham@duke.edu